Poster 890

Efficacy and Safety of Donidalorsen in Adolescent Patients With Hereditary Angioedema: A Subanalysis of the Phase 3 OASIS-HAE Study

Joshua Jacobs[1], Marc A Riedl[2], Laura Bordone[3], Kenneth B Newman[3], Sabrina Treadwell[3], Sophie Wang[3], Aaron Yarlas[3], Danny M Cohn[4]

[1]Allergy and Asthma Clinical Research, Inc., Walnut Creek, CA, USA; [2]Division of Allergy and Immunology, University of California San Diego School of Medicine, La Jolla, CA, USA; [3]Ionis, Carlsbad, CA, USA; [4]Department of Vascular Medicine, Amsterdam Cardiovascular Sciences, Amsterdam UMC, University of Amsterdam, Amsterdam, the Netherlands

Background

- Hereditary angioedema (HAE) is a rare disease characterized by frequently severe, potentially life-threatening attacks of tissue swelling[1,2]
- HAE is most often caused by C1 inhibitor (C1INH) deficiency (HAE-C1INH-Type1) or dysfunction (HAE-C1INH-Type2)[3]
- Symptoms usually first present in childhood, and adolescents experiencing HAE attacks report lower quality of life and elevated anxiety compared with healthy controls[4,5]
- Donidalorsen is an investigational RNA-targeted antisense oligonucleotide that specifically reduces prekallikrein production in the liver[6]
- In the phase 3 OASIS-HAE study (NCT05139810), donidalorsen 80 mg subcutaneously (SC) once every 4 weeks (Q4W) or once every 8 weeks (Q8W) significantly reduced HAE attack rate and improved patient-reported quality of life[6]
- Here, we report results for adolescent patients in OASIS-HAE

Study Design

Figure 1. Study Design

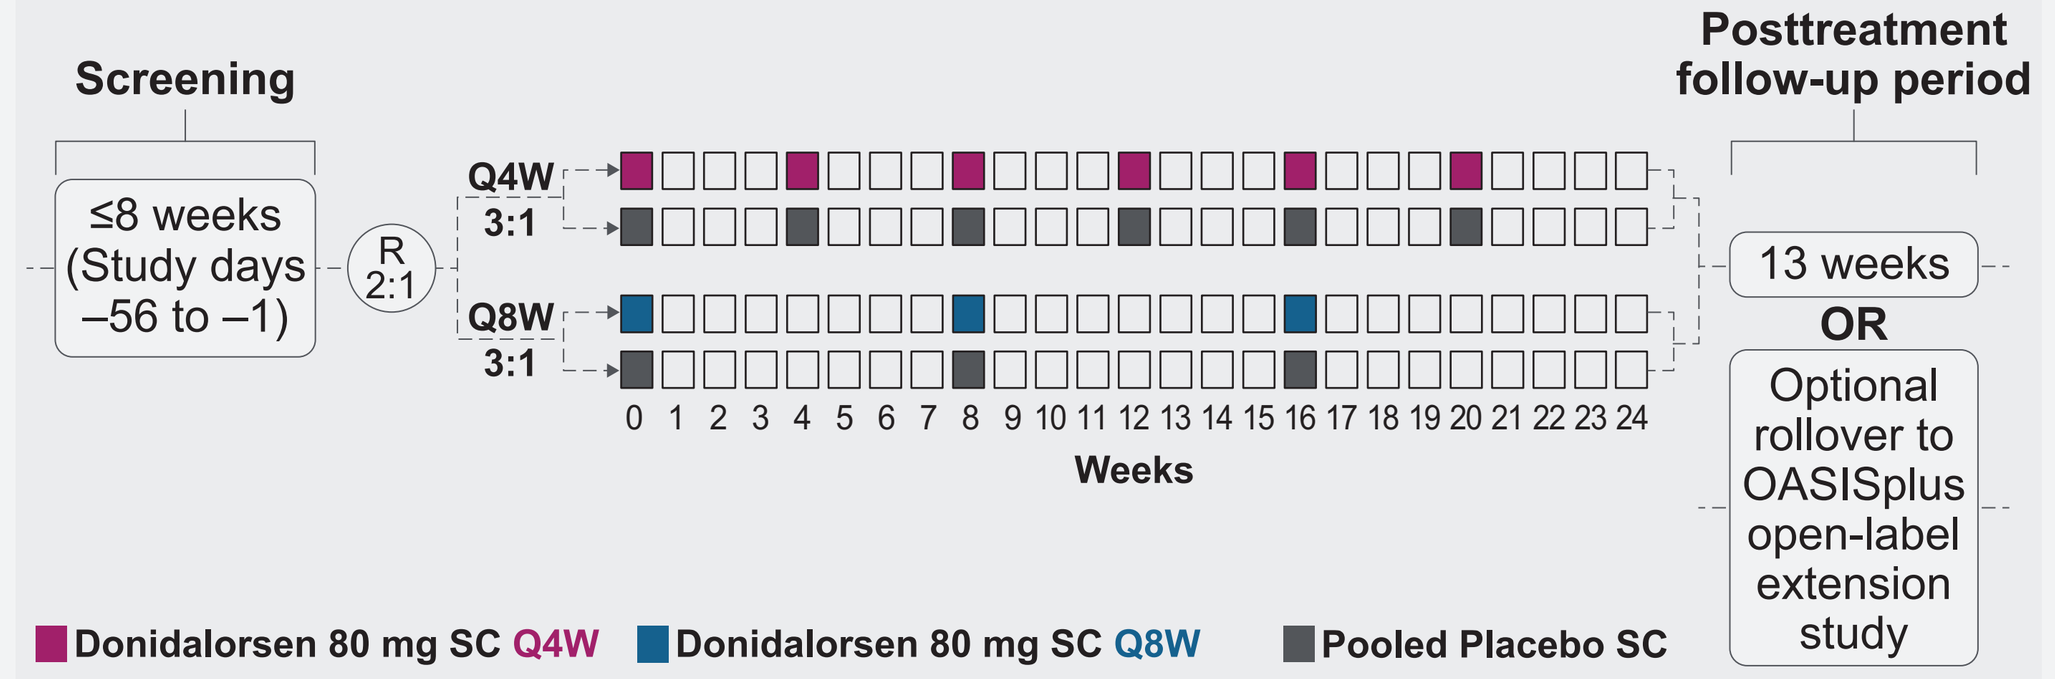

Screening
≤8 weeks (Study days −56 to −1)
R 2:1
Q4W 3:1
Q8W 3:1
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24
Weeks
Posttreatment follow-up period
13 weeks
OR
Optional rollover to OASISplus open-label extension study
Donidalorsen 80 mg SC Q4W
Donidalorsen 80 mg SC Q8W
Pooled Placebo SC

Q4W, once every 4 weeks; Q8W, once every 8 weeks; R, randomization; SC, subcutaneously.

- OASIS-HAE was a global, randomized, double-blind, placebo-controlled trial
- Patients were ≥12 years of age with HAE-C1INH-Type1 or HAE-C1INH-Type2
- Patients were randomized to receive donidalorsen 80 mg or placebo SC Q4W or Q8W for 24 weeks
- Endpoints included:
 - Time-normalized HAE attack rate
 - Incidence and severity of treatment-emergent adverse events (TEAEs)
 - Change in Angioedema Quality of Life Questionnaire (AE-QoL) total score (secondary endpoint)
 - Percent of patients with well-controlled disease based on the Angioedema Control Test (AECT; secondary endpoint)
 - Plasma prekallikrein concentrations (exploratory endpoint)

Results

Table 1. Demographics of Adolescent Patients

	Donidalorsen Q4W (n = 4)	Donidalorsen Q8W (n = 3)	Pooled Placebo (n = 0)	Total (N = 7)
Age, years, median (range)	15 (12–17)	15 (12–16)	—	15 (12–17)
Sex, n (%)				
Male	3 (75)	1 (33)	—	4 (57)
Female	1 (25)	2 (67)	—	3 (43)
Race, n (%)				
White	4 (100)	2 (67)	—	6 (86)
American Indian or Alaskan Native	0	1 (33)	—	1 (14)
Region, n (%)				
North America	0	2 (67)	—	2 (29)
Europe	1 (25)	0	—	1 (14)
Middle East	3 (75)	1 (33)	—	4 (57)

Q4W, once every 4 weeks; Q8W, once every 8 weeks.

- Seven adolescent patients (12–17 years of age) with HAE enrolled in OASIS-HAE
- No adolescent patients were randomized to placebo (randomization scheme did not stratify by age)

Figure 2. HAE Attack Rate in Adolescents (Weeks 0–24)

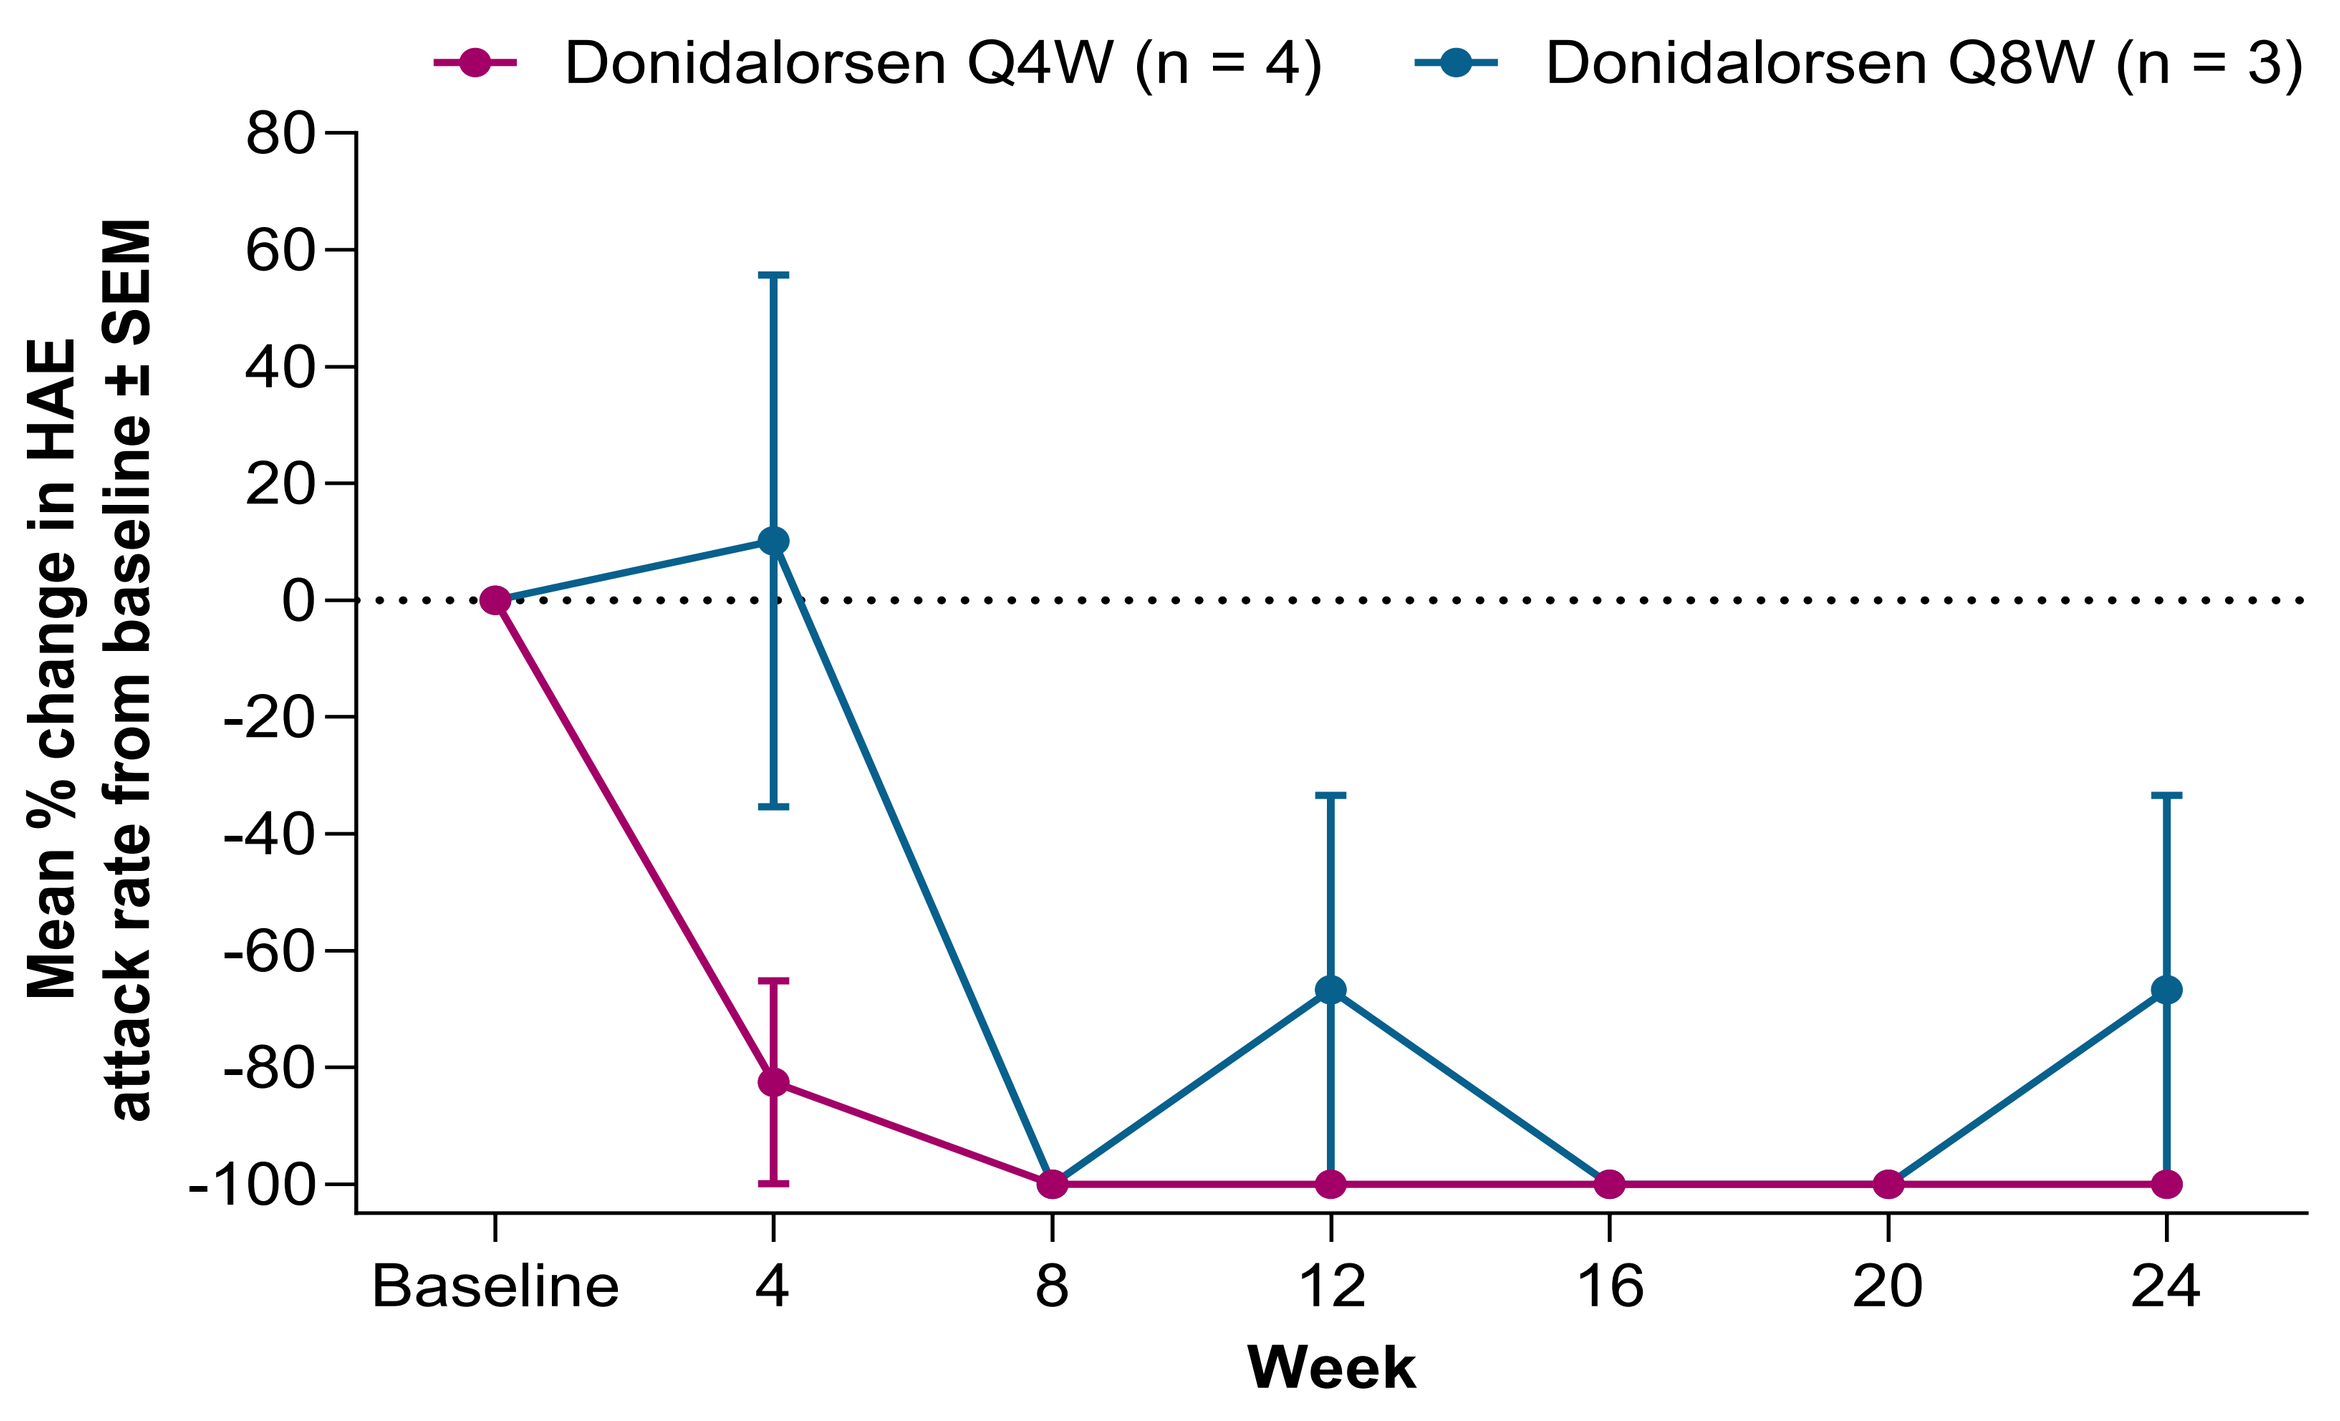

Donidalorsen Q4W (n = 4)
Donidalorsen Q8W (n = 3)
Mean % change in HAE attack rate from baseline ± SEM
80
60
40
20
0
-20
-40
-60
-80
-100
Baseline
4
8
12
16
20
24
Week

Baseline was defined as the 56-day run-in period prior to dosing in OASIS-HAE.
HAE, hereditary angioedema; Q4W, once every 4 weeks; Q8W, once every 8 weeks; SEM, standard error of the mean.

- Compared to baseline, mean HAE attack rates over Weeks 0 to 24 were 97% lower in the Q4W group and 71% lower in the Q8W group
 - Median reductions from baseline in HAE attack rates were −100% in the Q4W group and −87% in the Q8W group

Table 2. Summary of Adverse Events

	Donidalorsen Q4W (n = 4)	Donidalorsen Q8W (n = 3)	Total (N = 7)
Any TEAE,[a] n (%)	2 (50)	1 (33)	3 (43)
Related to study drug[b]	1[c] (25)	0	1[c] (14)
Leading to permanent treatment discontinuation	0	0	0
Any serious TEAE[a], n (%)	0	0	0
Severity of TEAEs[a], n (%)			
Mild	1 (25)	0	1 (14)
Moderate	1 (25)	1 (33)	2 (29)
Severe	0	0	0

[a]A TEAE is defined as any AE starting or getting worse on or after the first dose of the study drug. [b]Related is defined as related, possible, or missing relationship to study drug. [c]The TEAE was an increase in acne judged as possibly related to the study drug.
AE, adverse event; Q4W, once every 4 weeks; Q8W, once every 8 weeks; TEAE, treatment-emergent adverse event.

- Three adolescent patients had TEAEs, none of which were severe, serious, or led to treatment discontinuation
- The most common TEAE was nasopharyngitis

Figure 3. Patient-Reported Quality of Life (AE-QoL)

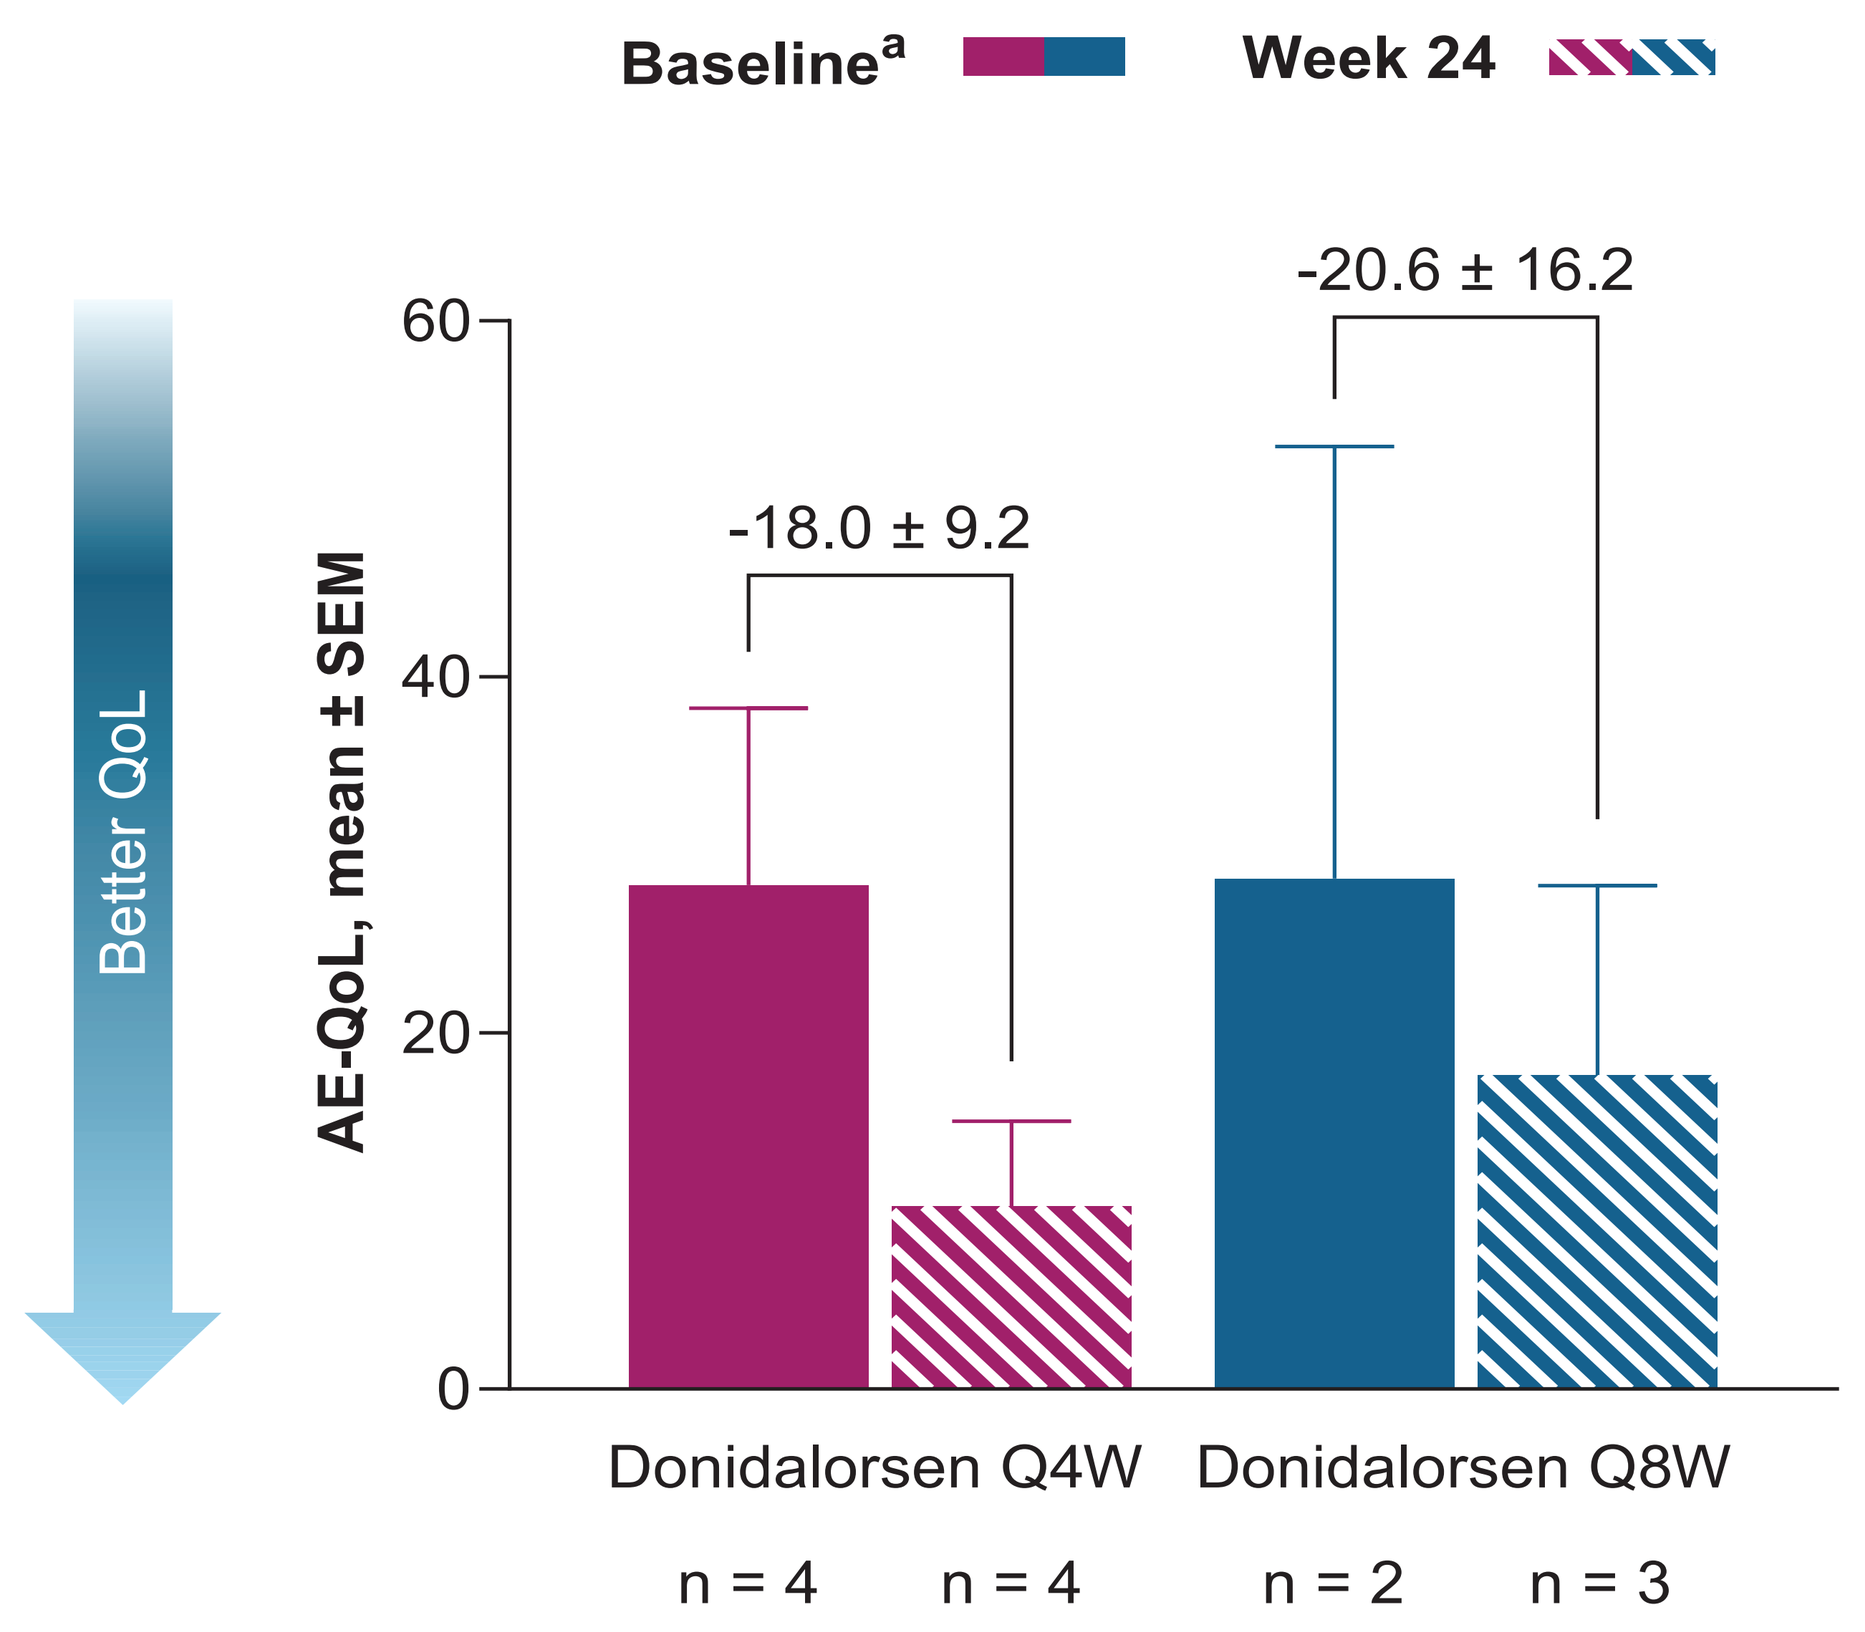

Baseline[a]
Week 24
Better QoL
AE-QoL, mean ± SEM
60
40
20
0
-18.0 ± 9.2
-20.6 ± 16.2
Donidalorsen Q4W
Donidalorsen Q8W
n = 4
n = 4
n = 2
n = 3

The AE-QoL was developed and validated in adult patients.[7]
[a]Defined as available AE-QoL total scores on study day 1.
AE-QoL, Angioedema Quality of Life Questionnaire; Q4W, once every 4 weeks; Q8W, once every 8 weeks; QoL, quality of life; SEM, standard error of the mean.

- Adolescents in both treatment groups reported clinically meaningful improvements in AE-QoL total score (≥6-point reduction)[8] at Week 24

Figure 4. Patients With Well-Controlled Disease (AECT)

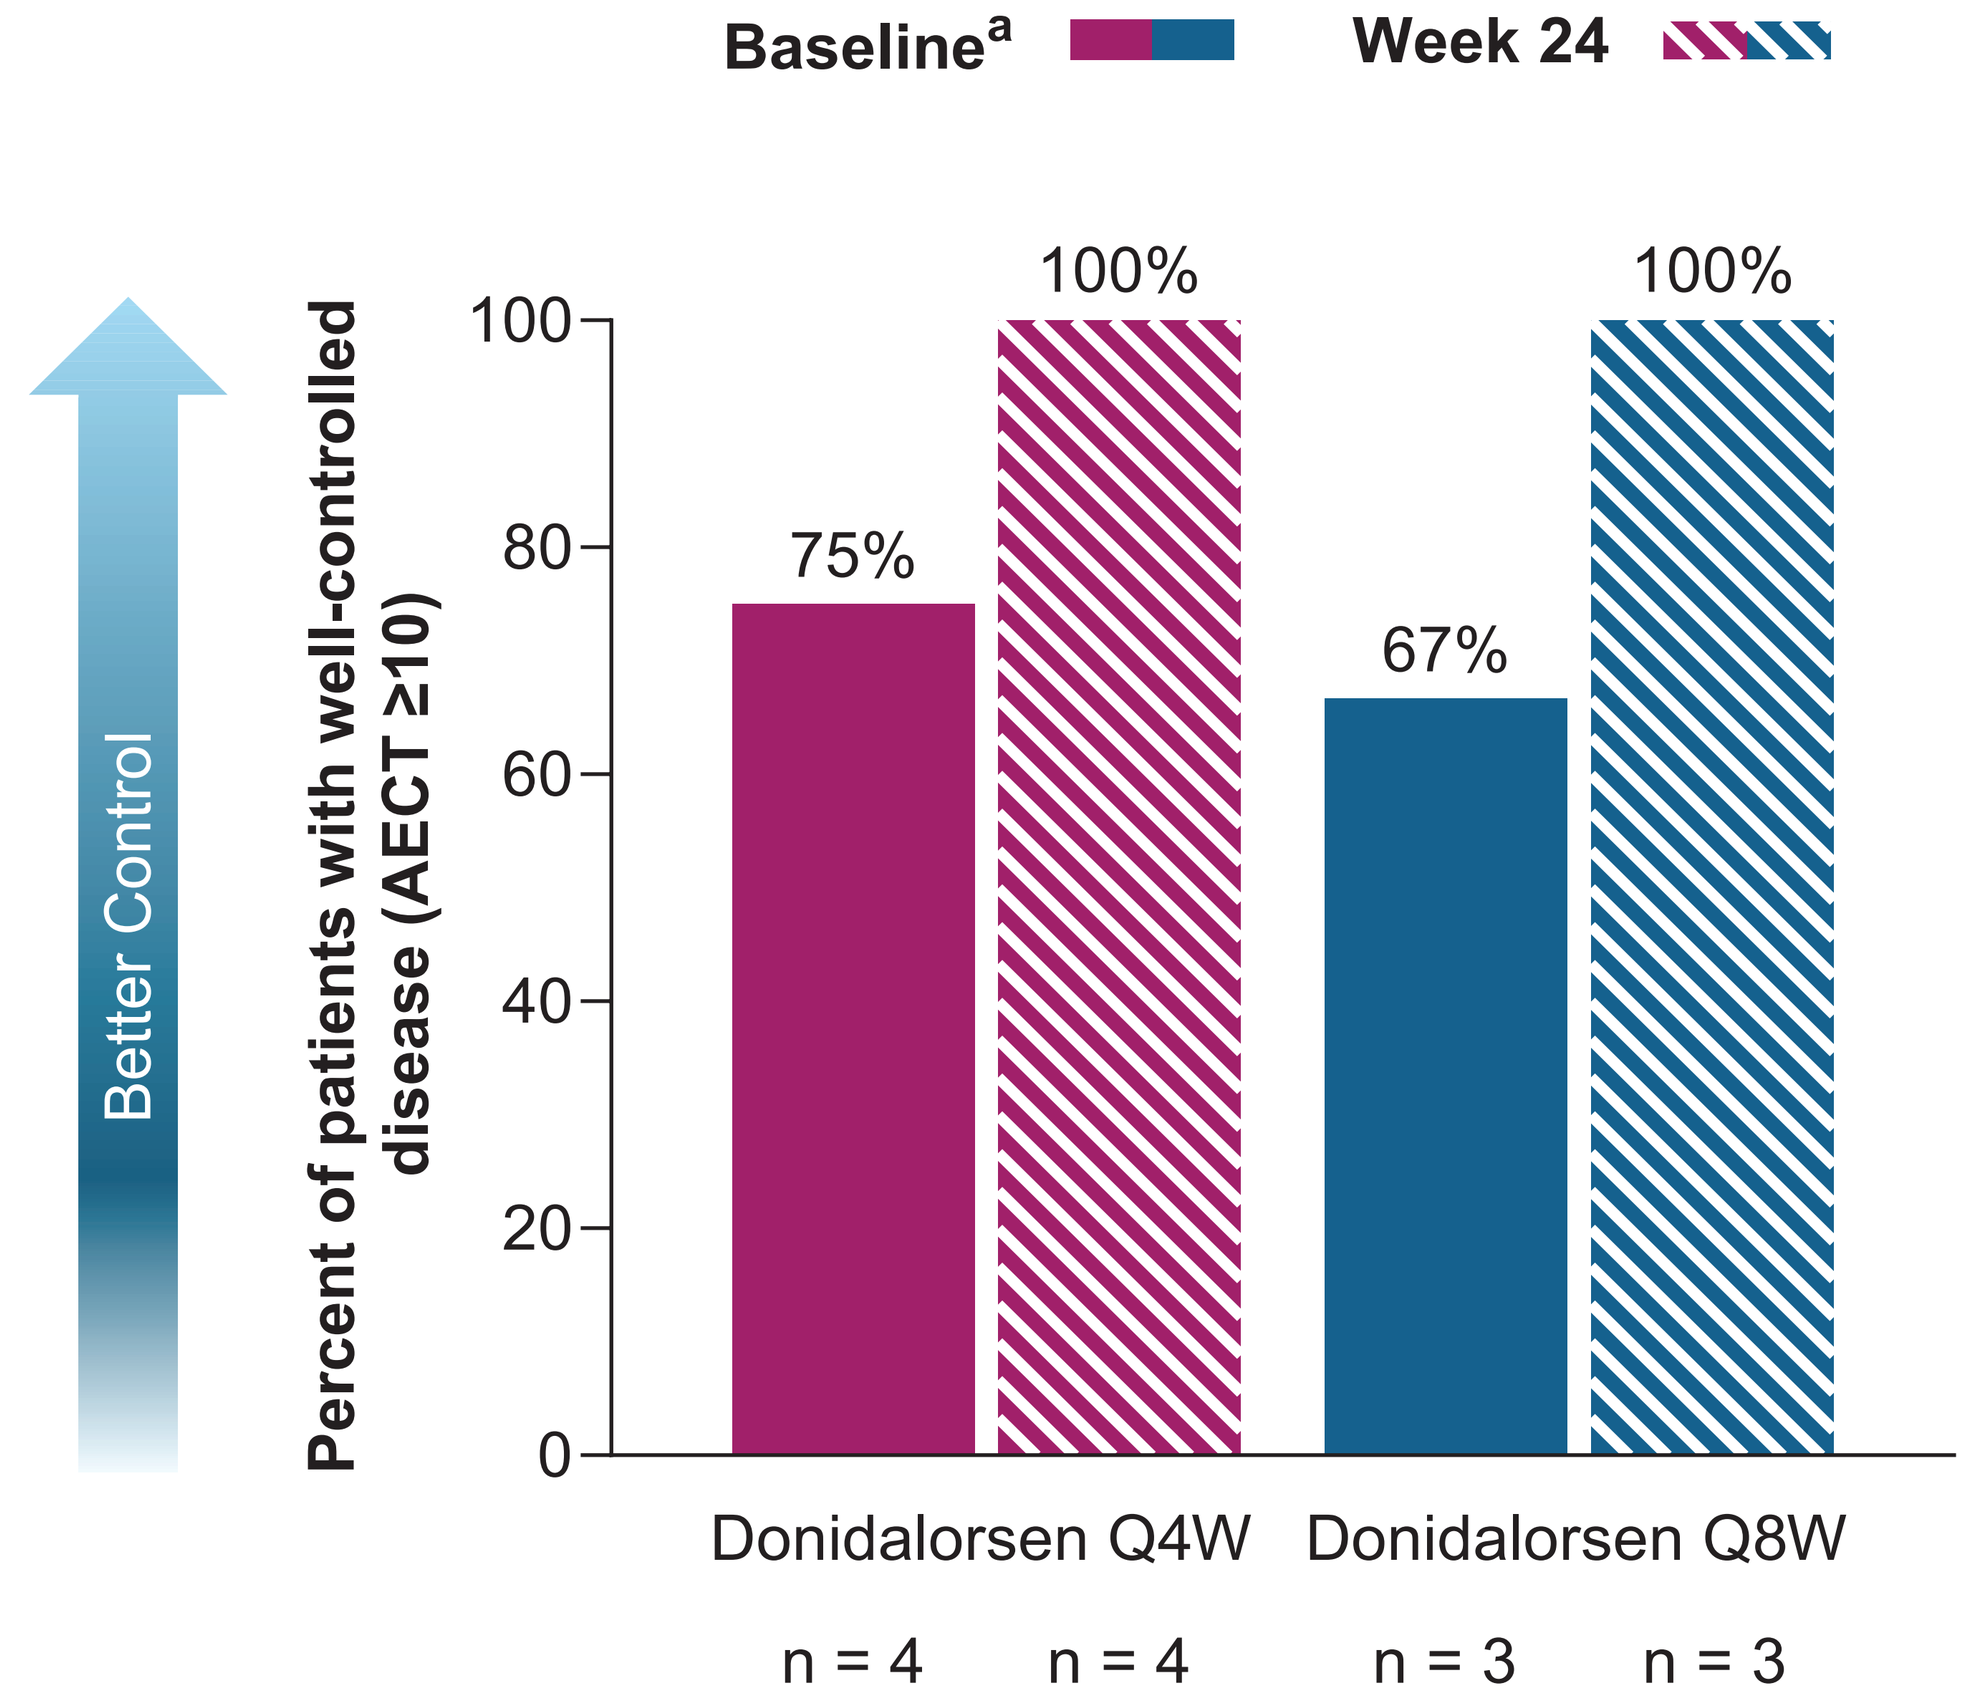

Baseline[a]
Week 24
Better Control
Percent of patients with well-controlled disease (AECT ≥10)
100
80
60
40
20
0
75%
100%
67%
100%
Donidalorsen Q4W
Donidalorsen Q8W
n = 4
n = 4
n = 3
n = 3

The AECT was developed and validated in adult patients.[9]
[a]Defined as AECT scores collected on study day 1.
AECT, Angioedema Control Test; Q4W, once every 4 weeks; Q8W, once every 8 weeks.

- By Week 24, all adolescent patients reported well-controlled disease (AECT ≥10)[10] on the AECT

Figure 5. Plasma Prekallikrein Concentrations in Adolescents (Weeks 0–24)

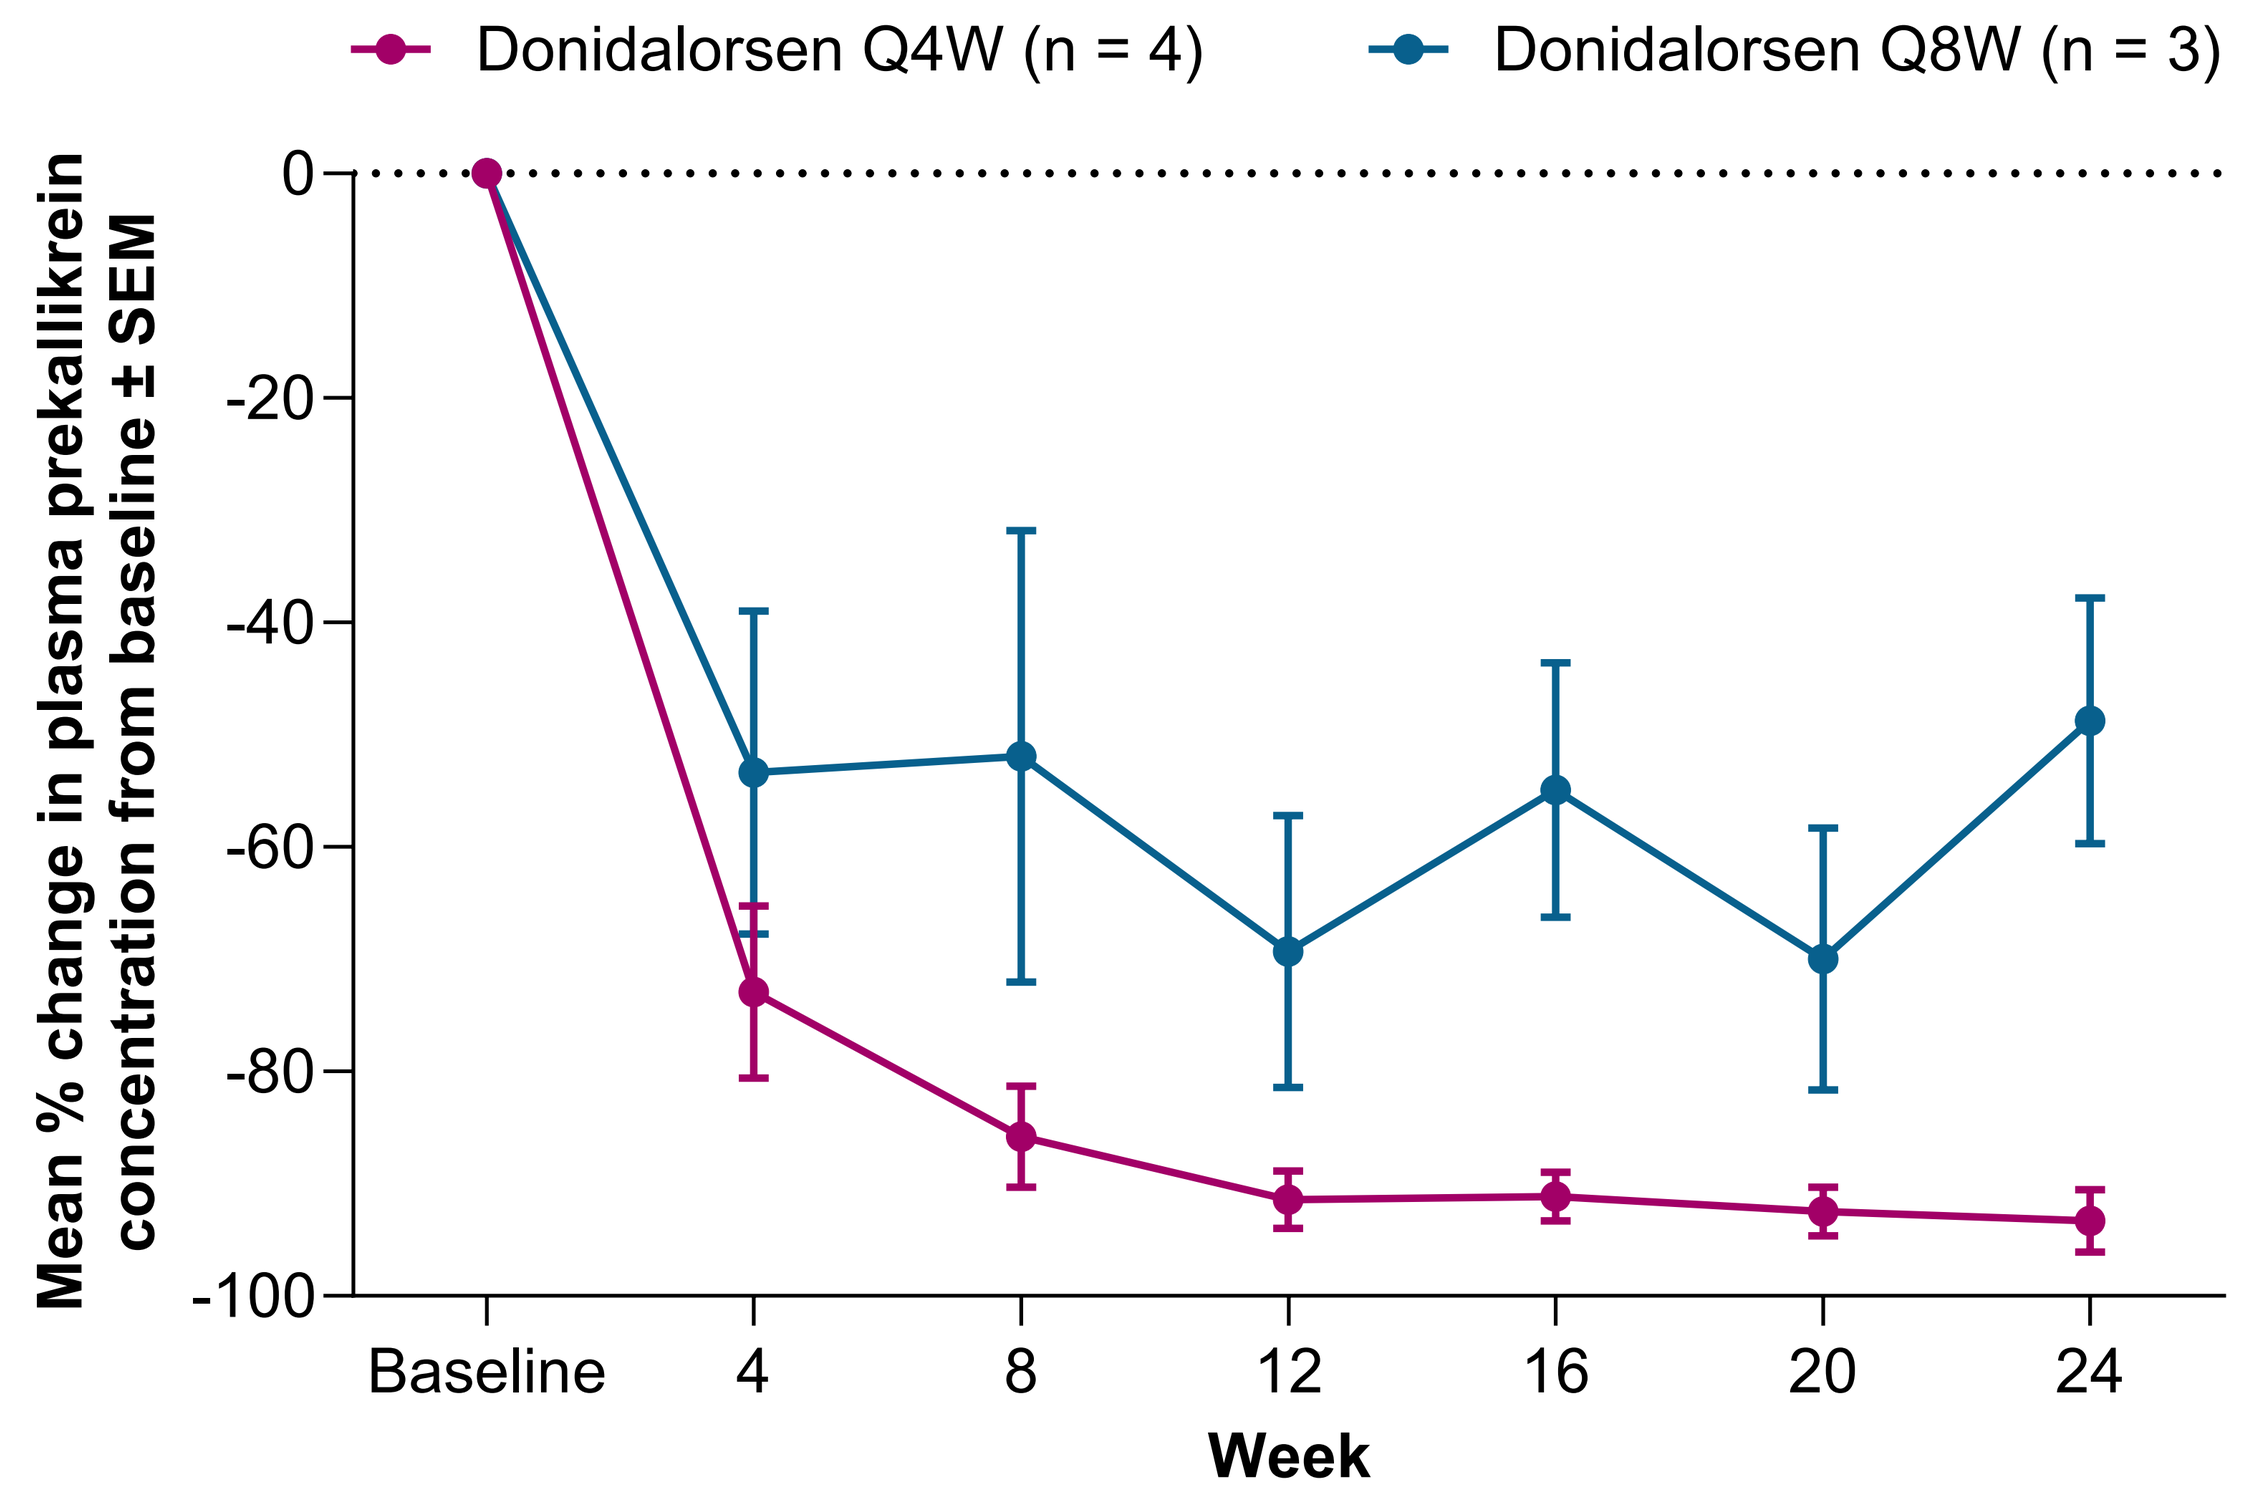

Donidalorsen Q4W (n = 4)
Donidalorsen Q8W (n = 3)
Mean % change in plasma prekallikrein concentration from baseline ± SEM
0
-20
-40
-60
-80
-100
Baseline
4
8
12
16
20
24
Week

Baseline was defined as the average of the pre-dose study day 1 value and the last pre-dose value prior to study day 1. If there was only 1 pre-dose measurement available, it was assigned as baseline.
Q4W, once every 4 weeks; Q8W, once every 8 weeks; SEM, standard error of the mean.

- Plasma prekallikrein concentrations decreased from baseline to Week 24 by 93% in the Q4W group and 49% in the Q8W group

Conclusions

HAE Attack Rate

- HAE attack rates in adolescent patients decreased from baseline by 97% in the Q4W group and 71% in the Q8W group over Weeks 0–24
- Improvements in HAE attack rates in adolescent patients were consistent with the overall efficacy results of OASIS-HAE[6]

Safety and Tolerability

- Donidalorsen had an acceptable safety profile in adolescents

Quality of Life and Disease Control

- Adolescent patients reported clinically meaningful improvements in quality of life, and all reported well-controlled disease by Week 24 of treatment

ACKNOWLEDGEMENTS

We thank the patients and their families, the trial site coordinators, and the OASIS-HAE team of investigators. We also thank Tracy Reigle, of Ionis, for graphic art assistance with the figures, and Nayna Sanathara, PhD, of Ionis, for editorial review. Medical writing and editorial assistance were provided by Jake Wilmot, PhD, of Red Nucleus, and funded by Ionis Pharmaceuticals, Inc. This study was funded by Ionis Pharmaceuticals, Inc.

DISCLOSURES

JJ has received research grants from Astria, BioCryst, CSL Behring, Pharvaris, and Takeda; consulted for BioCryst, CSL Behring, Pharvaris, and Takeda; and received honoraria for speaker presentations from BioCryst and Takeda. **MAR** has received research grants from BioCryst, BioMarin Pharmaceutical, CSL Behring, Ionis, KalVista, Pharvaris, and Takeda; consulted for Astria, BioCryst, BioMarin Pharmaceutical, Celldex, CSL Behring, Cycle Pharma, Grifols, Intellia Therapeutics, Ionis, KalVista, Pfizer, Pharming, Pharvaris, Sanofi-Regeneron, and Takeda; and provided speaker presentations for CSL Behring, Grifols, Pharming, and Takeda. **LB, KBN, ST, SW,** and **AY** are employees of Ionis and hold shares and/or options in Ionis. **DMC** has received speaker or consultancy fees from Astria, BioCryst, CSL Behring, Intellia, Ionis, KalVista, Pharvaris, and Takeda.

REFERENCES

1. Riedl MA, et al. *J Allergy Clin Immunol Pract*. 2024;12(4):911-8.
2. Raasch J, et al. *World Allergy Organ J*. 2023;16(6):100792.
3. Busse P, et al. *J Allergy Clin Immunol Pract*. 2022;10(3):716-22.
4. Engel-Yeger B, et al. *Pediatr Allergy Immunol*. 2017;28(4):370-6.
5. Kessel A, et al. *Pediatr Allergy Immunol*. 2017;28(7):692-8.
6. Riedl MA, et al. *N Engl J Med*. 2024;391(1):21-31.
7. Weller K, et al. *Allergy*. 2012;67:1289-98
8. Weller K, et al. *Allergy*. 2016;71(8):1203-9.
9. Weller K, et al. *Allergy*. 2020;75(5):1165–77
10. Weller K, et al. *J Allergy Clin Immunol Pract*. 2020;8(6):2050-57.e4.

American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Congress • San Diego, CA, USA • February 28–March 3, 2025 • Poster 890

jsjacobsmd@sbcglobal.net