Poster 699

Participant-Reported Ease-of-Use With a Prefilled, Single-Dose, Disposable Autoinjector for the Treatment of Hereditary Angioedema

Hetal Khatri[1], Phillip Estepa[1], Brendan Cummings[1]

[1]Ionis, Carlsbad, CA, USA

Background

Hereditary Angioedema

- Hereditary angioedema (HAE) is a rare disease characterized by attacks of tissue swelling that are often severe and can be life-threatening[1]
- Donidalorsen is an investigational RNA-targeted antisense oligonucleotide that specifically reduces prekallikrein production in the liver[2]
- The pivotal phase 3 OASIS-HAE study showed that donidalorsen 80 mg subcutaneously once every 4 weeks significantly reduced HAE attack rate and had an acceptable safety profile[2]

Human Factors Testing

- A single-dose, disposable autoinjector is being evaluated for use to administer donidalorsen
- For combination products, the US Food and Drug Administration recommends human factors (HF) testing to evaluate risk analysis of the product's user interface
- **The main objective of this HF validation study was to confirm that the donidalorsen autoinjector could be used by both patients and caregivers at home without repeated patterns of use errors that would result in harm**

Study Design

Figure 1. Study Design of Human Factors Testing

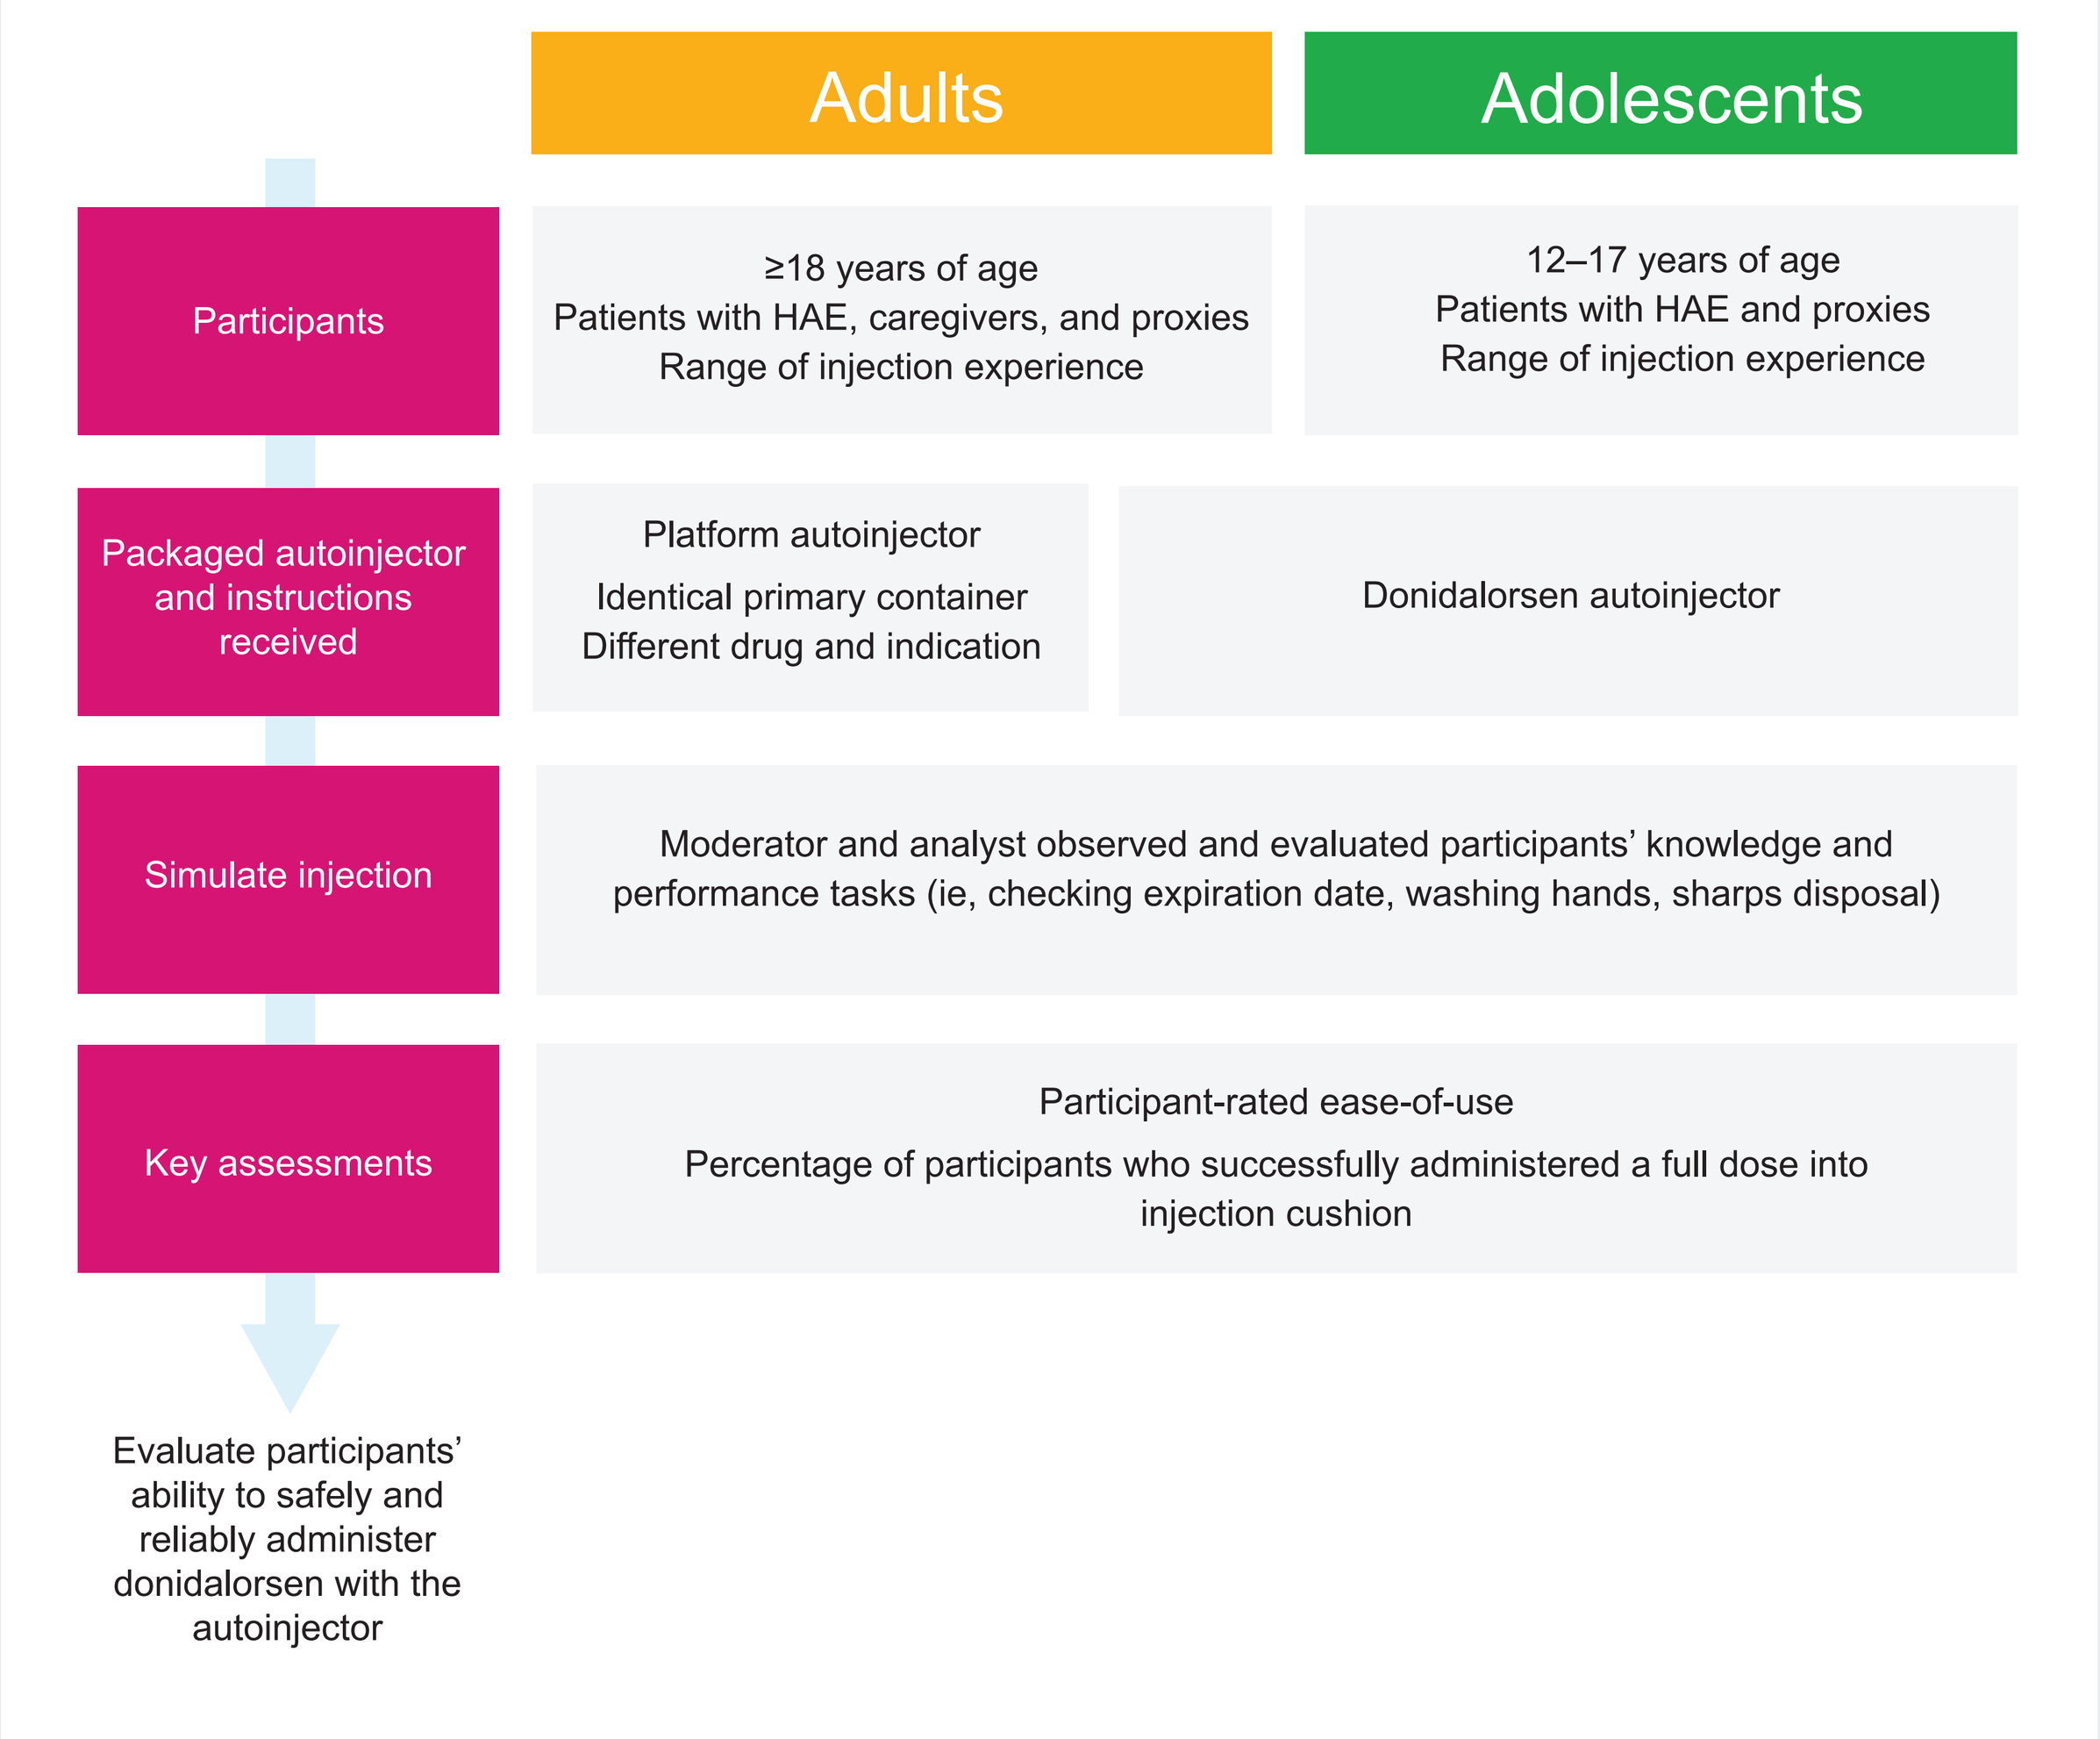

Adults
Adolescents
Participants
≥18 years of age
Patients with HAE, caregivers, and proxies
Range of injection experience
12–17 years of age
Patients with HAE and proxies
Range of injection experience
Packaged autoinjector and instructions received
Platform autoinjector
Identical primary container
Different drug and indication
Donidalorsen autoinjector
Simulate injection
Moderator and analyst observed and evaluated participants' knowledge and performance tasks (ie, checking expiration date, washing hands, sharps disposal)
Key assessments
Participant-rated ease-of-use
Percentage of participants who successfully administered a full dose into injection cushion
Evaluate participants' ability to safely and reliably administer donidalorsen with the autoinjector

HAE, hereditary angioedema.

Results

Table 1. Demographics of Adult Participants (Donidalorsen HF Validation)

	Patients (n = 16)	Caregivers (n = 15)
Age, years, mean (range)	50 (21–72)	53 (32–74)
Sex, n (%)		
Female	11 (69)	7 (47)
Male	5 (31)	8 (53)
Highest education, n (%)		
High school	4 (25)	2 (13)
Some college	4 (25)	5 (33)
College	8 (50)	2 (13)
Advanced degree	0	4 (27)
Trade school	0	2 (13)
Patients with reported impairments, n (%)*		
Dexterity	1 (6)	3 (20)
Visual	13 (81)	12 (80)
Hearing	1 (6)	0 (0)

*Dexterity impairments included arthritis, parkinsonism, mild joint pain, and Dupuytren's syndrome; visual impairments included corrected vision (distance and/or reading) and unspecified impairment; hearing impairments included hearing aid use. HF, human factors.

- A total of 31 adult patients with HAE, proxy patients, caregivers of patients with HAE, and proxy caregivers attempted to administer donidalorsen with the autoinjector by following the included "Instructions for Use"
- Individuals had a range of injection experience, including some with no prior experience

Table 2. Demographics of Adolescent Participants (Donidalorsen HF Validation)

	Adolescent patients (n = 21)
Age, years, mean (range)	15 (12–17)
Sex, n (%)	
Female	13 (62)
Male	8 (38)
School year, n (%)	
7th grade (approximately 12–13 years of age)	3 (14)
8th grade (approximately 13–14 years of age)	3 (14)
9th grade (approximately 14–15 years of age)	5 (24)
10th grade (approximately 15–16 years of age)	3 (14)
11th grade (approximately 16–17 years of age)	3 (14)
12th grade (approximately 17–18 years of age)	4 (19)
Patients with reported impairments, n (%)*	
Dexterity	2 (10)
Visual	6 (29)
Hearing	0 (0)

*Dexterity impairments included numbness and occasional swelling in hands from HAE flare-ups; visual impairments included corrected vision (distance and/or reading) and single-eye blindness. HF, human factors.

- A total of 21 adolescents between the ages of 12 and 17 participated, comprised of both patients with HAE and proxy patients
- Individuals had a range of injection experience, including some with no prior experience

Figure 2. Ease-of-Use Ratings (Donidalorsen HF Validation Participants)

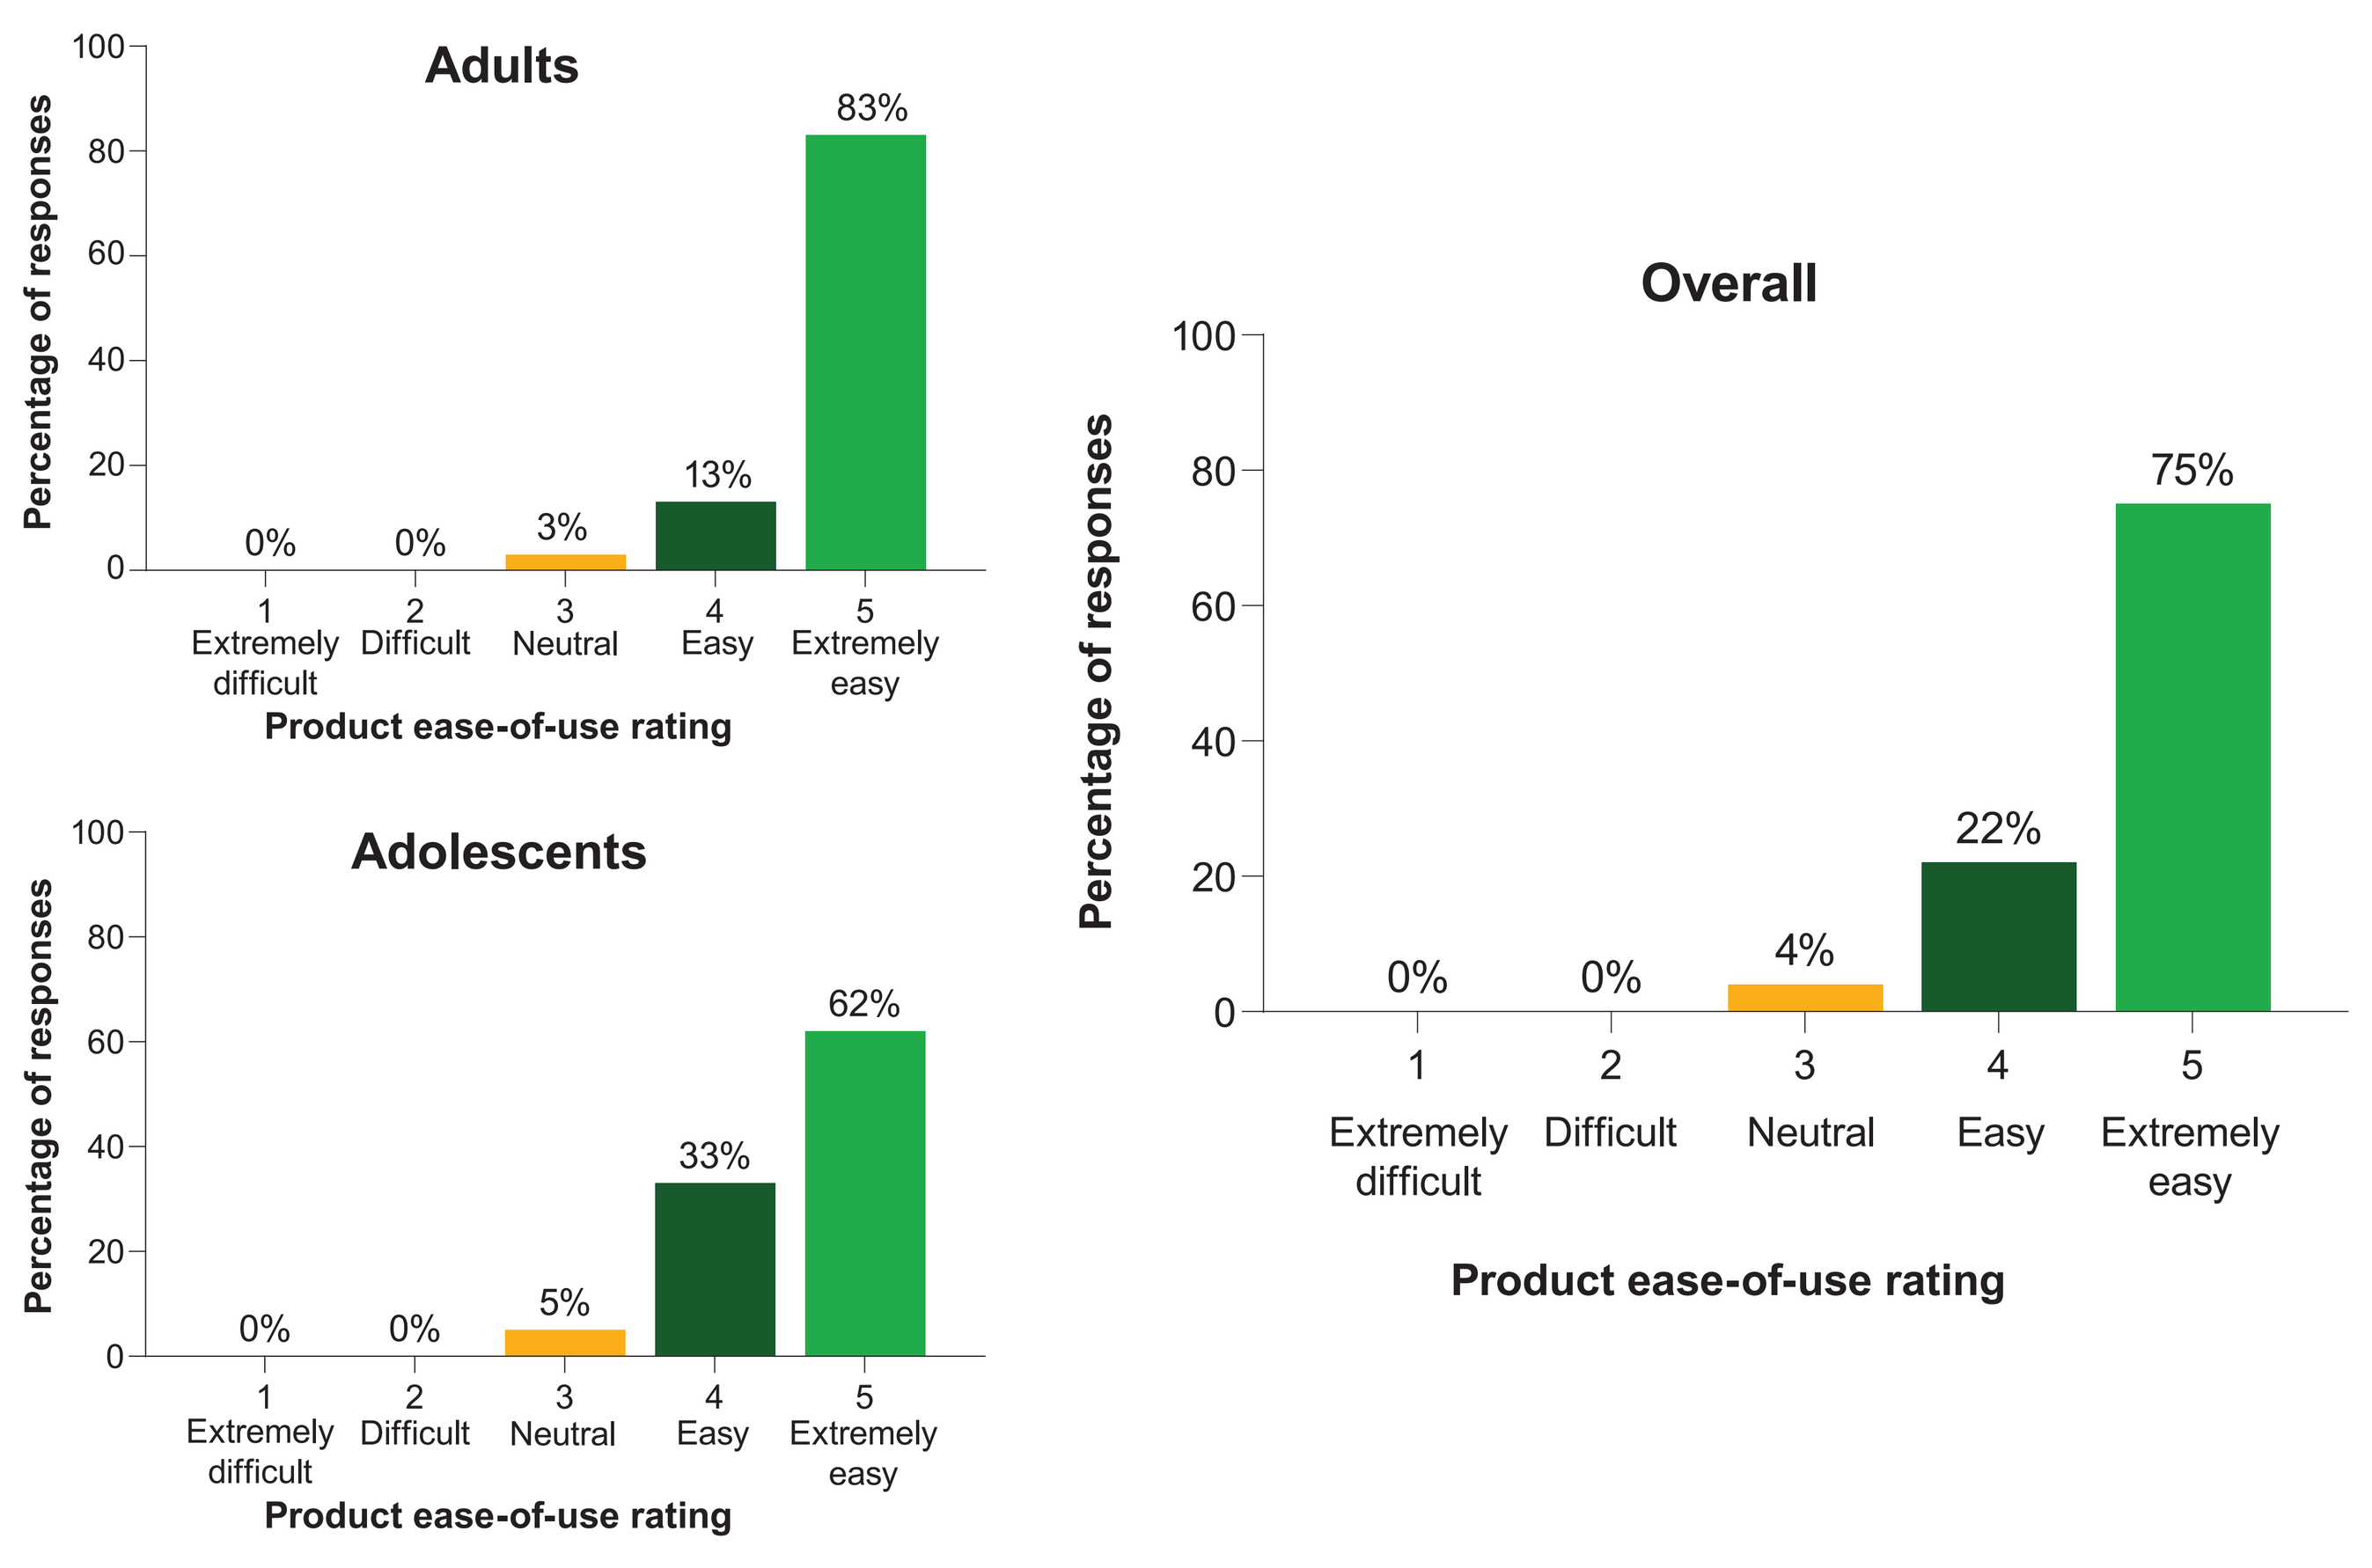

Adults
Adolescents
Overall
Percentage of responses
0% 0% 3% 13% 83%
0% 0% 5% 33% 62%
0% 0% 4% 22% 75%
1 Extremely difficult
2 Difficult
3 Neutral
4 Easy
5 Extremely easy
Product ease-of-use rating

HF, human factors.

- The majority of participants in both the adult and adolescent validation studies rated the autoinjector as "extremely easy" to use

Table 3. Demographics of Platform Study Participants

	Patients (n = 31)	Caregivers (n = 31)
Age, years, mean (range)	61 (39–75)	56 (32–81)
Sex, n (%)		
Female	22 (71)	19 (61)
Male	9 (29)	12 (39)
Highest education, n (%)		
High school	3 (10)	2 (6)
Some college	12 (39)	10 (32)
College	11 (35)	11 (35)
Advanced degree	5 (16)	8 (26)
Patients with reported impairments, n (%)*		
Dexterity	31 (100)	3 (10)
Visual	31 (100)	25 (81)
Hearing	10 (32)	4 (13)

*Dexterity impairments included arthritis, carpal tunnel, neuropathy, tendinitis, and Raynaud's disease; visual impairments included corrected vision (distance and/or reading), eye floaters, cataracts, retinopathy, astigmatism, glaucoma, and macular degeneration; hearing impairments included hearing loss, tinnitus, and use of hearing aid. Patients participating in the platform study had relatively high rates of dexterity, visual, and hearing impairments due to the platform product's disease indications.

- The platform study with a platform drug product that uses an identical primary container and autoinjector for a different indication included the following:
 – 16 injection-experienced and 15 injection-naïve adult proxy patients
 – 15 injection-experienced and 16 injection-naïve adult lay proxy caregivers

Figure 3. Ease-of-Use Ratings in Platform Study vs Donidalorsen Autoinjector

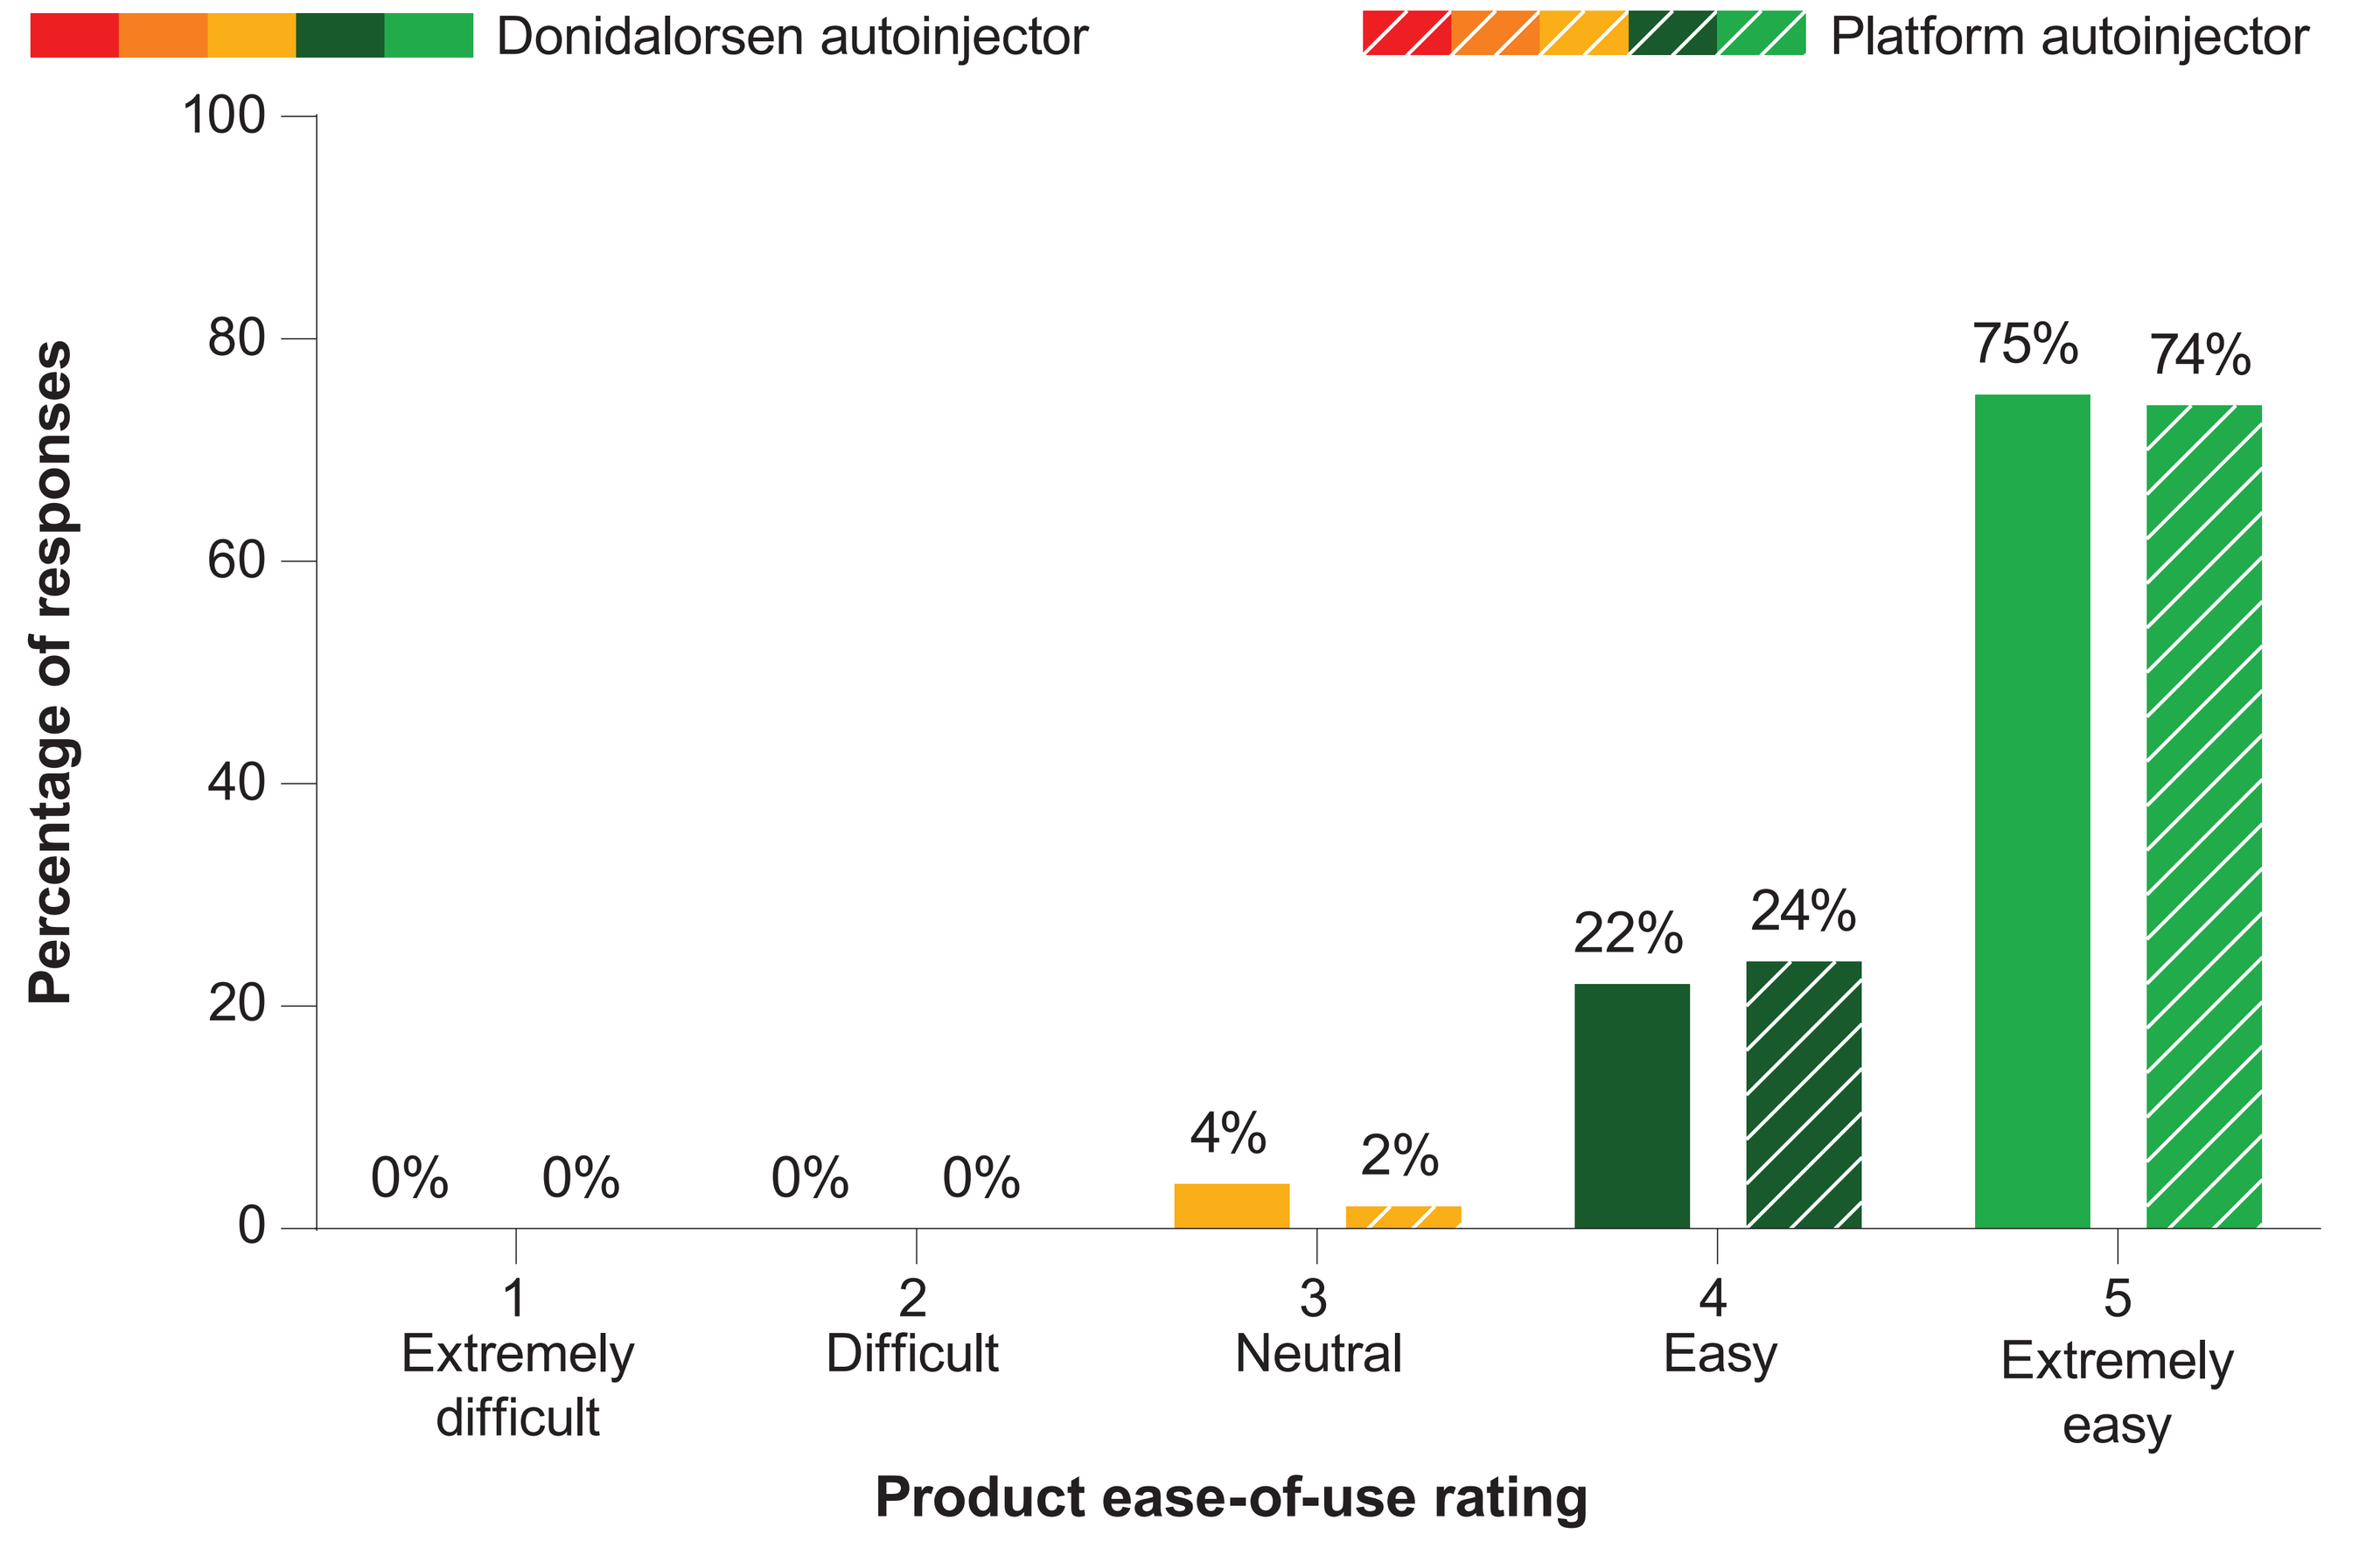

Donidalorsen autoinjector
Platform autoinjector
Percentage of responses
0% 0% 0% 0% 4% 2% 22% 24% 75% 74%
1 Extremely difficult
2 Difficult
3 Neutral
4 Easy
5 Extremely easy
Product ease-of-use rating

- Ease-of-use ratings were similar between the platform drug product and the donidalorsen autoinjector

Figure 4. Successful Administration Performed in HF Studies

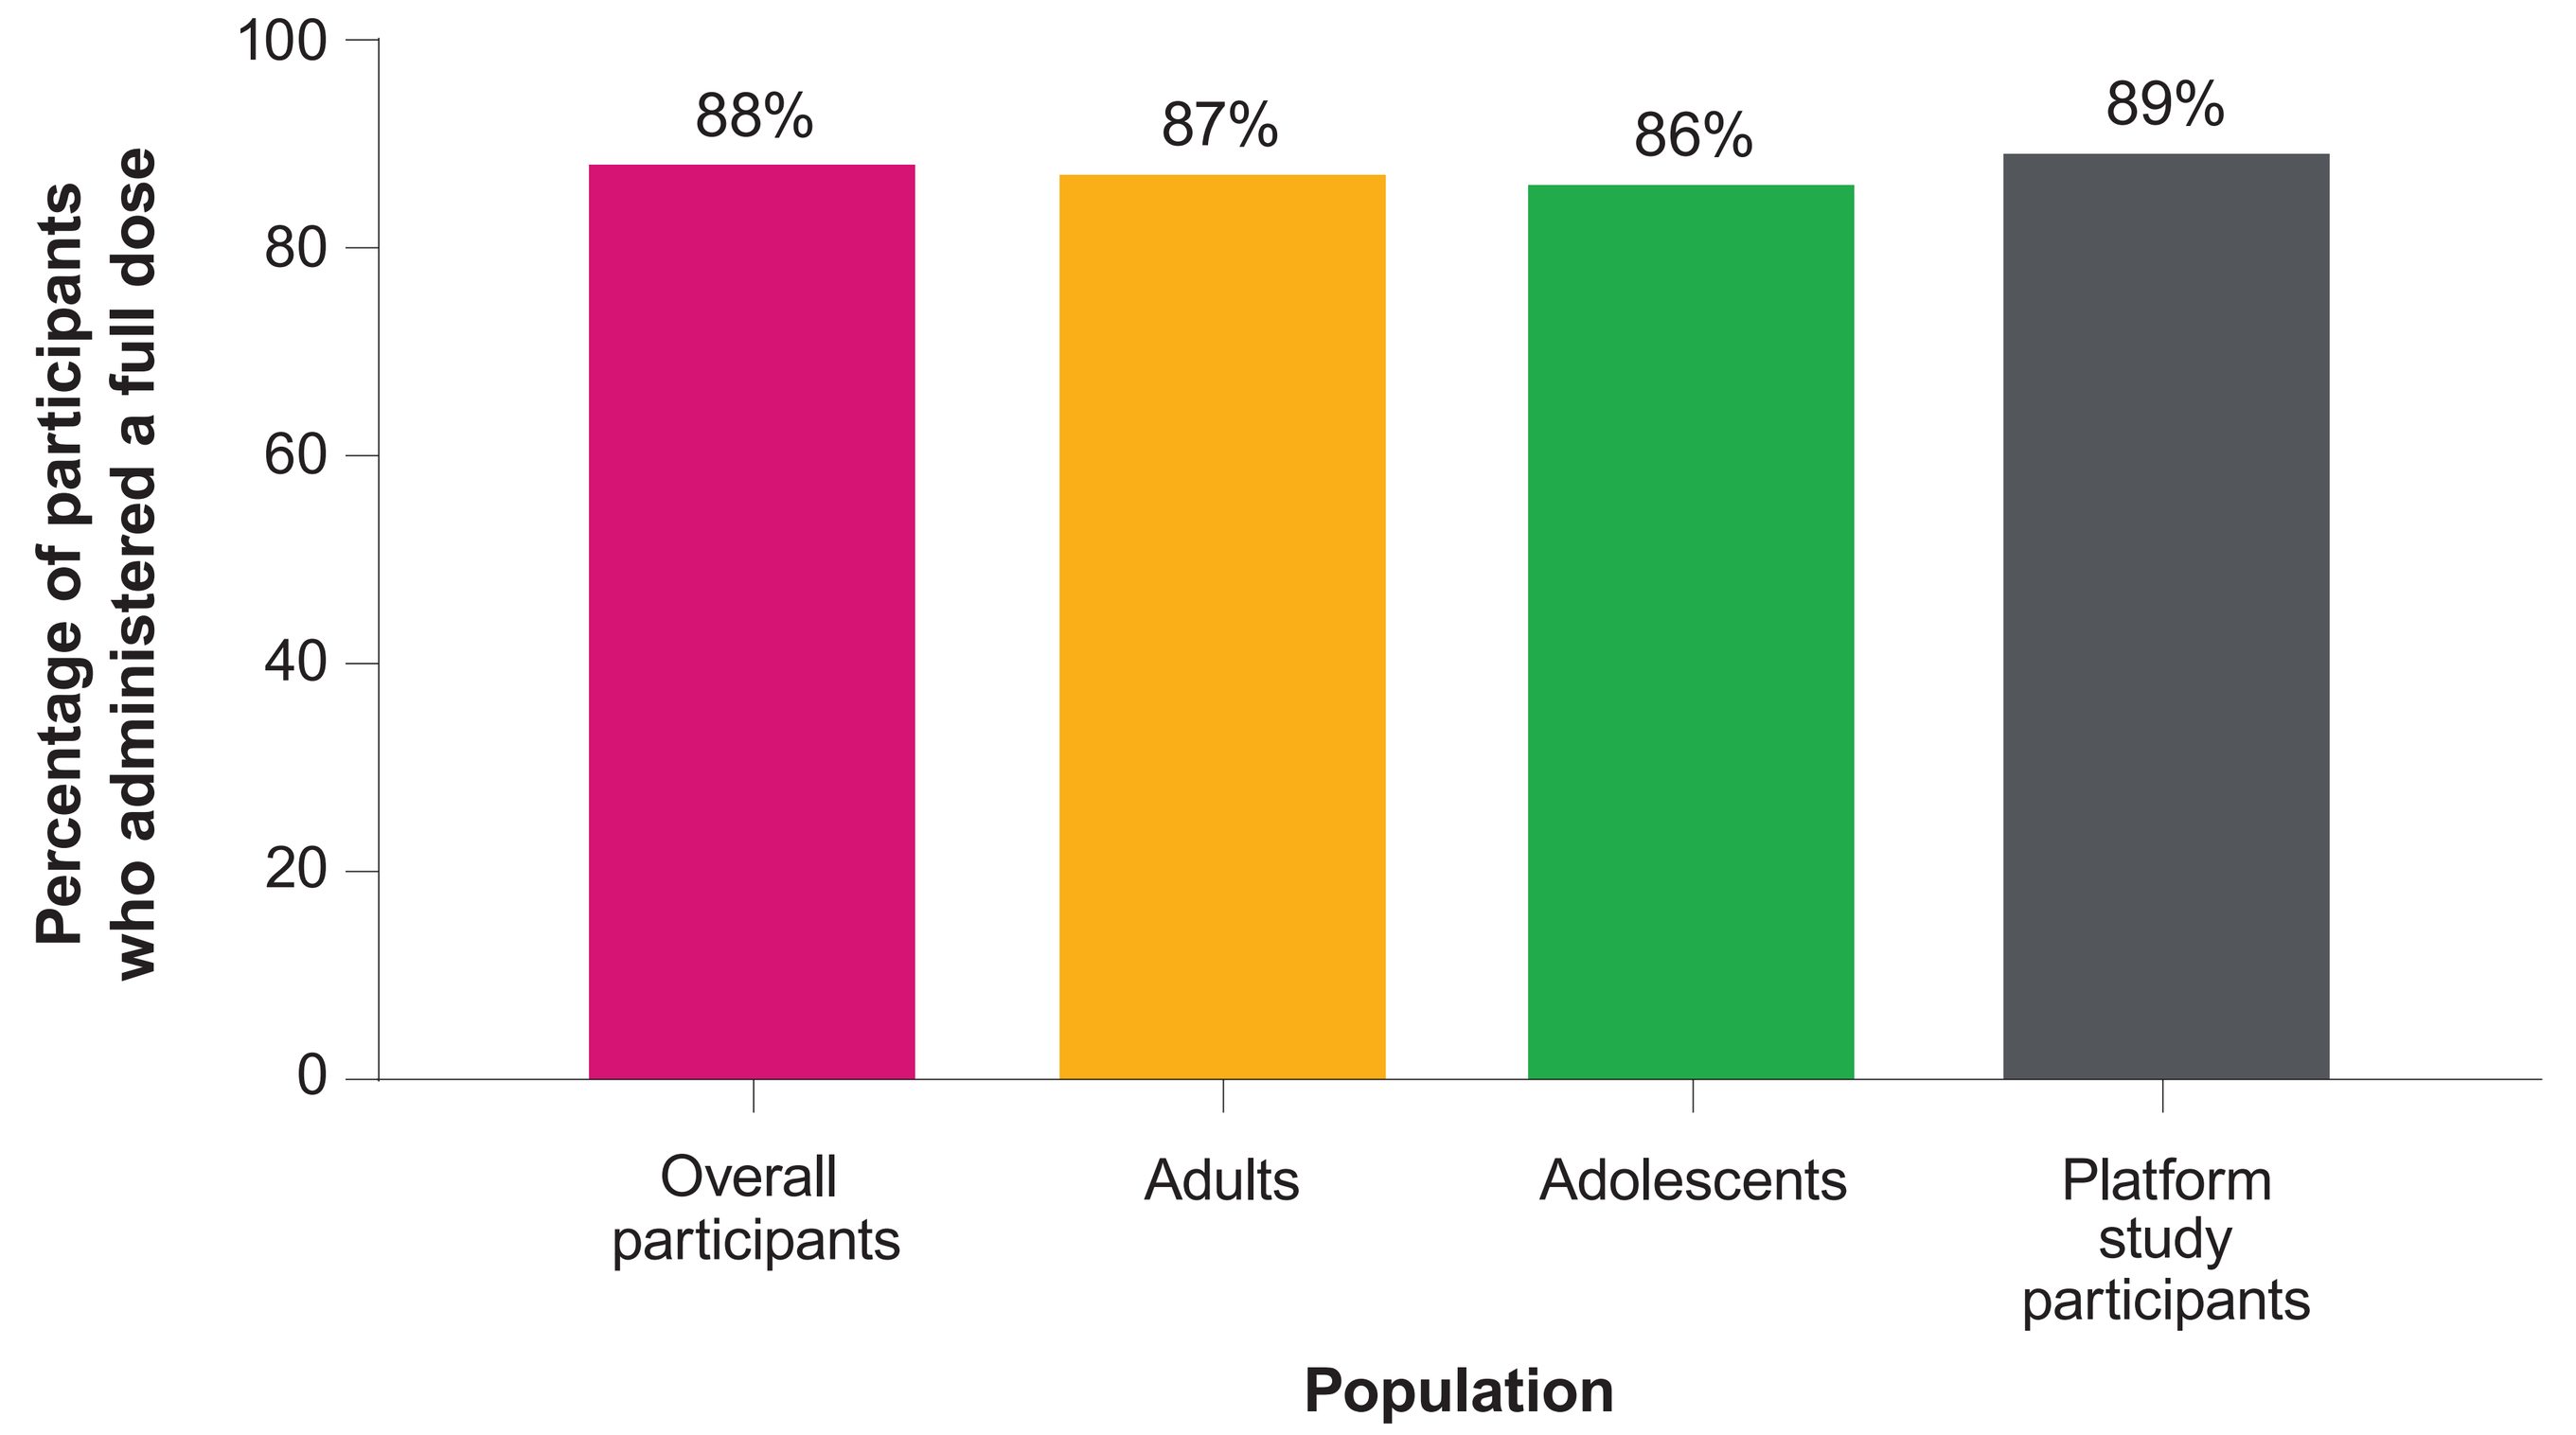

Percentage of participants who administered a full dose
88% 87% 86% 89%
Overall participants
Adults
Adolescents
Platform study participants
Population

HF, human factors.

- Despite participants receiving no direction other than the "Instructions for Use" document, injection administration success rates were high across all HF validation cohorts
- The most common reason for a failed injection in the adult and adolescent groups was leaking at the injection site due to an insufficient amount of time hof ageing the autoinjector at the injection site

Conclusions

HF testing validates use of the donidalorsen autoinjector as a delivery system for its intended users, uses, and use environments

Success Rate

- Overall, 88% of study participants in the HF studies were able to administer a full autoinjector dose into an injection cushion, with no instruction beyond the included "Instructions for Use" document

Patient Experience

- Regardless of prior injection experience, most patients found the autoinjector "easy" or "extremely easy" to use, with none reporting use as "difficult" or "extremely difficult"

ACKNOWLEDGMENTS

We thank the participants and their families, the site coordinators, and the team of investigators. We also thank Tracy Reigle, of Ionis, for graphic art assistance with the figures and Nayna Sanathara, PhD, of Ionis, for editorial review. Medical writing and editorial assistance were provided by Red Nucleus and funded by Ionis Pharmaceuticals, Inc. Human factors validation testing was conducted by a third-party firm, Emergo by UL, and funded by Ionis Pharmaceuticals, Inc.

DISCLOSURES

HK, PE, and **BC** are employees of Ionis.

REFERENCES

1. Riedl MA, et al. *J Allergy Clin Immunol Pract*. 2024;12(4):911-8.
2. Riedl MA, et al. *N Engl J Med*. 2024;391(1):21-31.

American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Congress • San Diego, CA, USA • February 28–March 3, 2025 • Poster 699

HKhatri@ionis.com