IMAGINATION: A Global Phase 3 Trial of RO7434656, an Antisense Oligonucleotide Inhibitor of Complement Factor B, in IgA Nephropathy

SA-PO885

Jonathan Barratt,[1] Jürgen Flöge,[2] Vishal Duggal,[3] Nadine Schmit,[4] Ji Cheng,[5] Jeannette Lo,[3] Brad H. Rovin[6]

[1]Department of Cardiovascular Sciences, University of Leicester, Leicester, UK; [2]Division of Nephrology, University Hospital, Rheinisch-Westfälische Technische Hochschule (RWTH) Aachen, Aachen, Germany; [3]Genentech, Inc., South San Francisco, CA, USA; [4]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [5]Hoffmann-La Roche Ltd, Mississauga, ON, Canada; [6]Department of Internal Medicine, The Ohio State University College of Medicine, Columbus, OH, USA

BACKGROUND

- IgA nephropathy (IgAN) is a chronic, progressive autoimmune disease and is the most common cause of primary glomerulonephritis worldwide[1,2]
 - Approximately 25% to 30% of patients with IgAN develop kidney failure within 20 to 25 years of presentation[3]
- Current treatments include blood pressure control through inhibition of the renin-angiotensin system and lifestyle modifications[3]
- IgAN is a multi-hit disease in which immune complexes of galactose-deficient IgA1 and associated autoantibodies accumulate in the kidney and trigger activation of the alternative complement pathway[1]
 - Factor B is a key protease in the activation and amplification of the alternative complement pathway, and serum levels of factor B are increased in patients with IgAN[4]
 - Overactivation of the alternative complement pathway has been implicated in the pathogenesis and severity of IgAN[5]
- RO7434656 (IONIS-FB-LRx; ISIS 696844), a ligand-conjugated antisense oligonucleotide targeting complement factor B messenger RNA (mRNA), was engineered for enhanced delivery to the liver as the primary site of factor B production (**Figure 1**)
 - RO7434656 binds to and destroys complement factor B mRNA, preventing production of factor B protein
 - In an initial data cut (July 2022) of a Phase 2 trial (NCT04014335), RO7434656 inhibited alternative complement pathway activation and demonstrated a clinically meaningful reduction in the urine protein-to-creatinine ratio (UPCR; **Figure 2**) and stabilization of the estimated glomerular filtration rate (eGFR) in patients with IgAN[6]
 - RO7434656 was generally well-tolerated, with no serious adverse events

OBJECTIVE

- To evaluate the efficacy, safety and pharmacokinetics of RO7434656 compared with those of placebo in adults with biopsy-confirmed primary IgAN

METHODS

Study design and participant population

- IMAGINATION (NCT05797610) is a Phase 3, randomized, double-blind, placebo-controlled trial (**Figure 3**)
- Participants will be randomized 1:1 to either RO7434656 or matching placebo administered as a subcutaneous injection every 4 weeks for 105 weeks
 - After 105 weeks, participants will have the opportunity to continue double-blind treatment or switch to open label treatment
 - After the first 8 doses of the study treatment, administration may be completed by the participant or another caregiver at their home or other suitable location upon completion of training
- The study will be conducted at approximately 225 sites globally (**Figure 4**)
- Key inclusion and exclusion criteria are shown in **Table 1**
- Primary, secondary and exploratory endpoints are shown in **Table 2**

Statistical analysis

- The sample size was calculated to provide approximately 90% power and detect a 30% difference in the primary endpoint between RO7434656 and placebo groups at a 2-sided 0.025 (α) significance level
- The primary analysis is based on Week 37 assessments and updated analyses on Week 105 assessments

CONCLUSIONS

- IMAGINATION aims to evaluate the efficacy and safety of RO7434656 in adults with IgAN using a broad range of assessments over 105 weeks
- The unique antisense modality and long tissue half-life of RO7434656 enables monthly subcutaneous administration to inhibit the alternative complement pathway

RO7434656 AND THE IMAGINATION TRIAL

Figure 1. Mechanism of Action of RO7434656

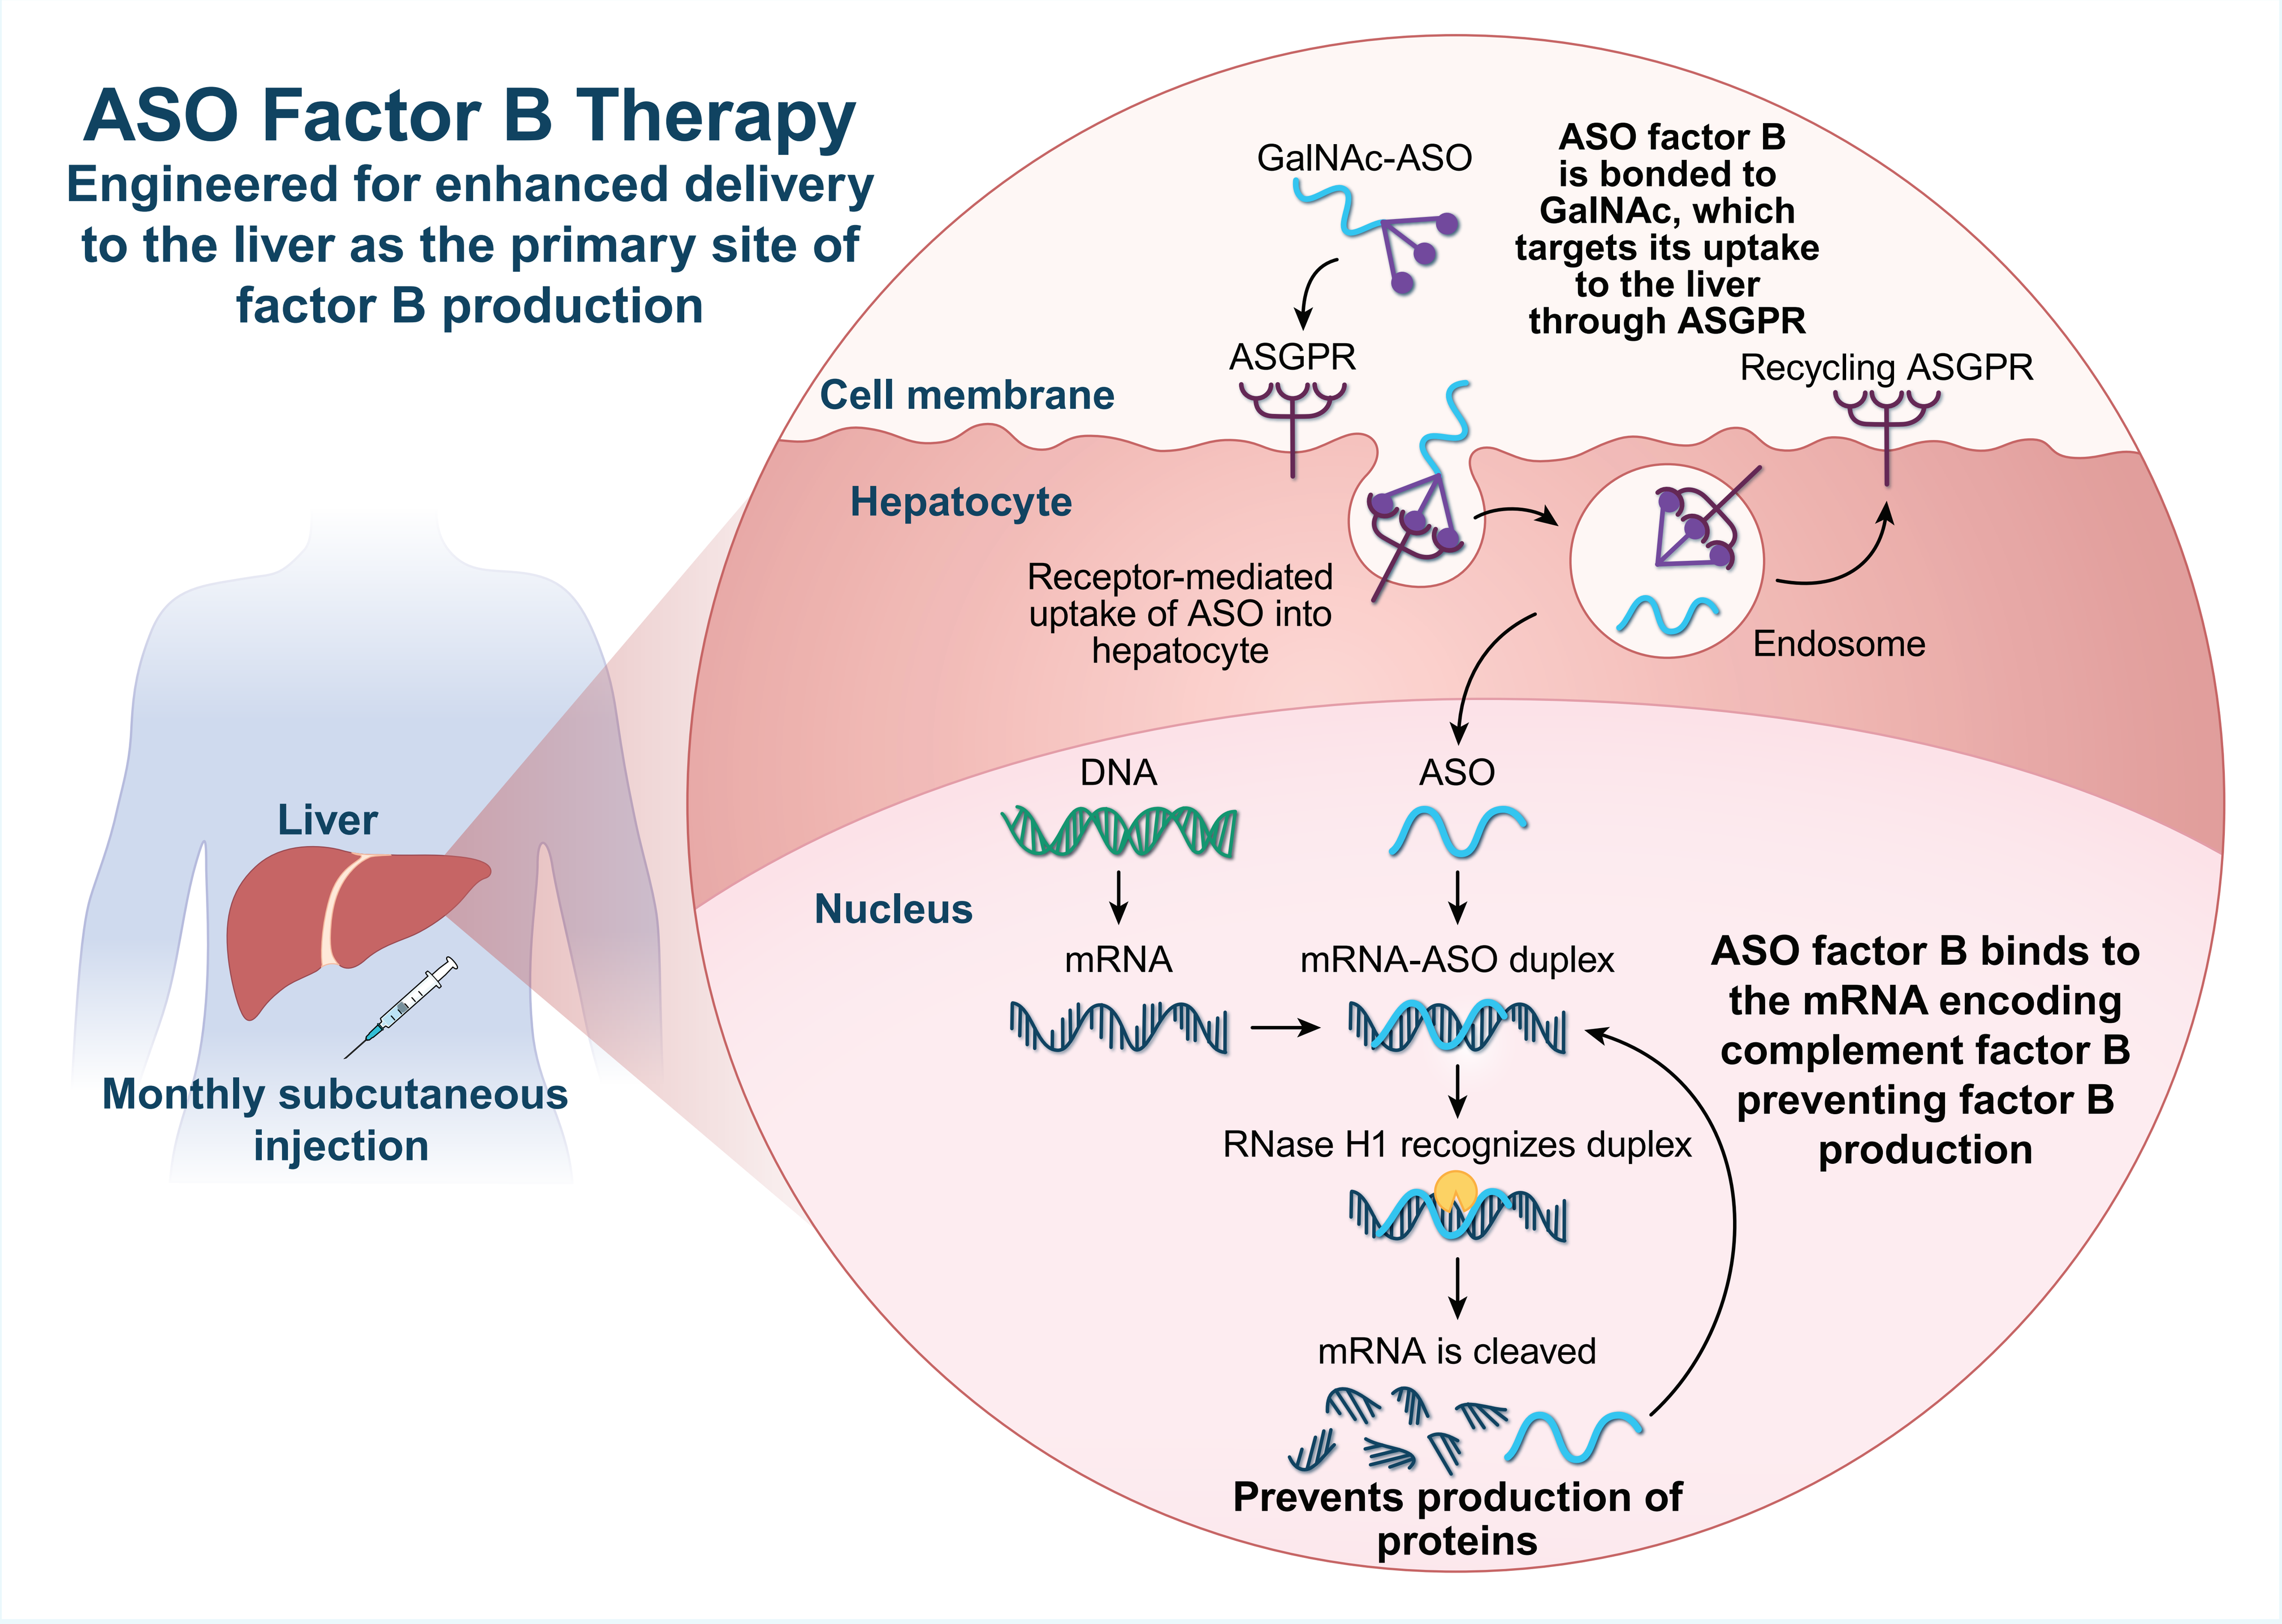

ASO Factor B Therapy
Engineered for enhanced delivery to the liver as the primary site of factor B production
Liver
Monthly subcutaneous injection
GalNAc-ASO
ASO factor B is bonded to GalNAc, which targets its uptake to the liver through ASGPR
ASGPR
Cell membrane
Recycling ASGPR
Hepatocyte
Receptor-mediated uptake of ASO into hepatocyte
Endosome
DNA
ASO
Nucleus
mRNA
mRNA-ASO duplex
ASO factor B binds to the mRNA encoding complement factor B preventing factor B production
RNase H1 recognizes duplex
mRNA is cleaved
Prevents production of proteins

ASO, antisense oligonucleotide; ASGPR, asialoglycoprotein receptor; GalNac, N-acetyl-d-galactosamine; mRNA, messenger RNA; RNase, ribonuclease.

Figure 2. Change in (**A**) Factor B, (**B**) Key Complement Parameters and (**C**) UPCR From an Initial Data Cut (July 2022) of a Phase 2 Trial of RO7434656 (NCT04014335)[6]* in Patients With IgAN (n=10)

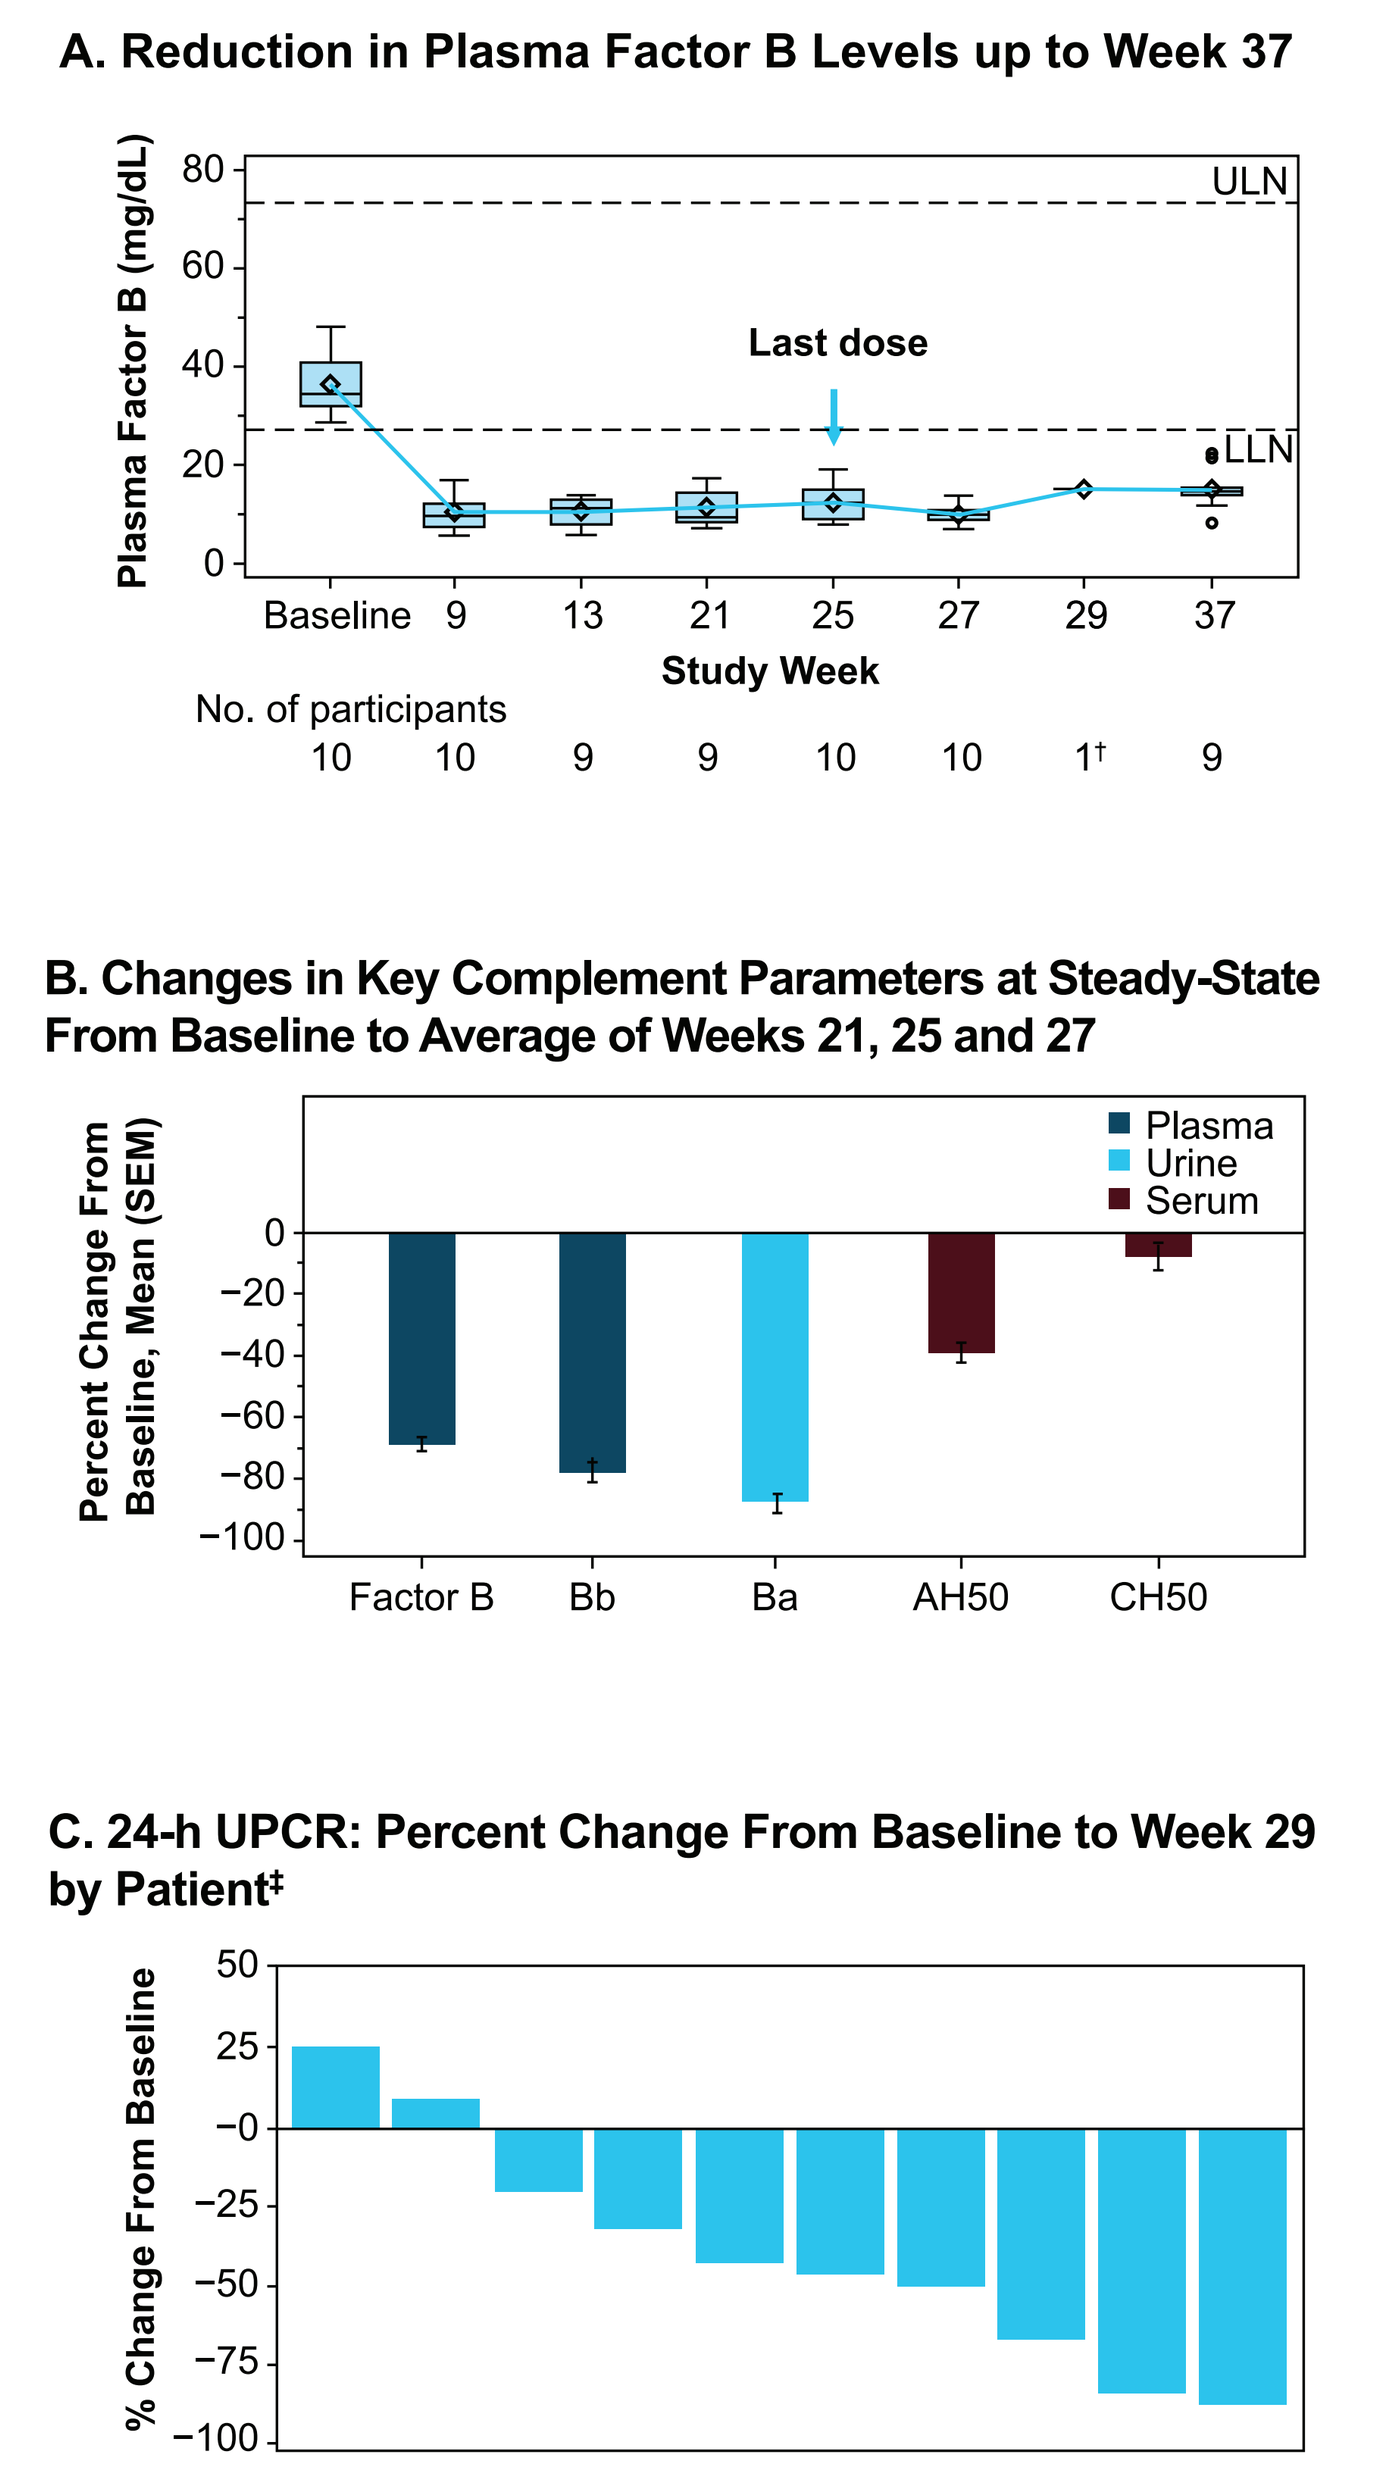

A. Reduction in Plasma Factor B Levels up to Week 37
Plasma Factor B (mg/dL)
80
60
40
20
0
ULN
LLN
Last dose
Baseline 9 13 21 25 27 29 37
Study Week
No. of participants
10 10 9 9 10 10 1† 9
B. Changes in Key Complement Parameters at Steady-State From Baseline to Average of Weeks 21, 25 and 27
Percent Change From Baseline, Mean (SEM)
Plasma
Urine
Serum
0 −20 −40 −60 −80 −100
Factor B Bb Ba AH50 CH50
C. 24-h UPCR: Percent Change From Baseline to Week 29 by Patient‡
% Change From Baseline
50 25 −0 −25 −50 −75 −100

Barbour S, et al. Presented at: American Society of Nephrology (ASN) Kidney Week 2022; November 3-6, 2022; Orlando, FL[7]. Poster SA-P0714. Images reprinted with permission by the author.
IgAN, IgA nephropathy; LLN, lower limit of normal; ULN, upper limit of normal; UPCR, urine protein-to-creatinine ratio.
*Study sponsor is Ionis Pharmaceuticals, Inc. Patients received subcutaneous injections of RO7434656 70 mg at Weeks 1, 3, 5, and every 4 weeks thereafter.
†Additional complement sampling time points following protocol amendment.

Figure 3. Study Design of the Phase 3 IMAGINATION Trial (NCT05797610)

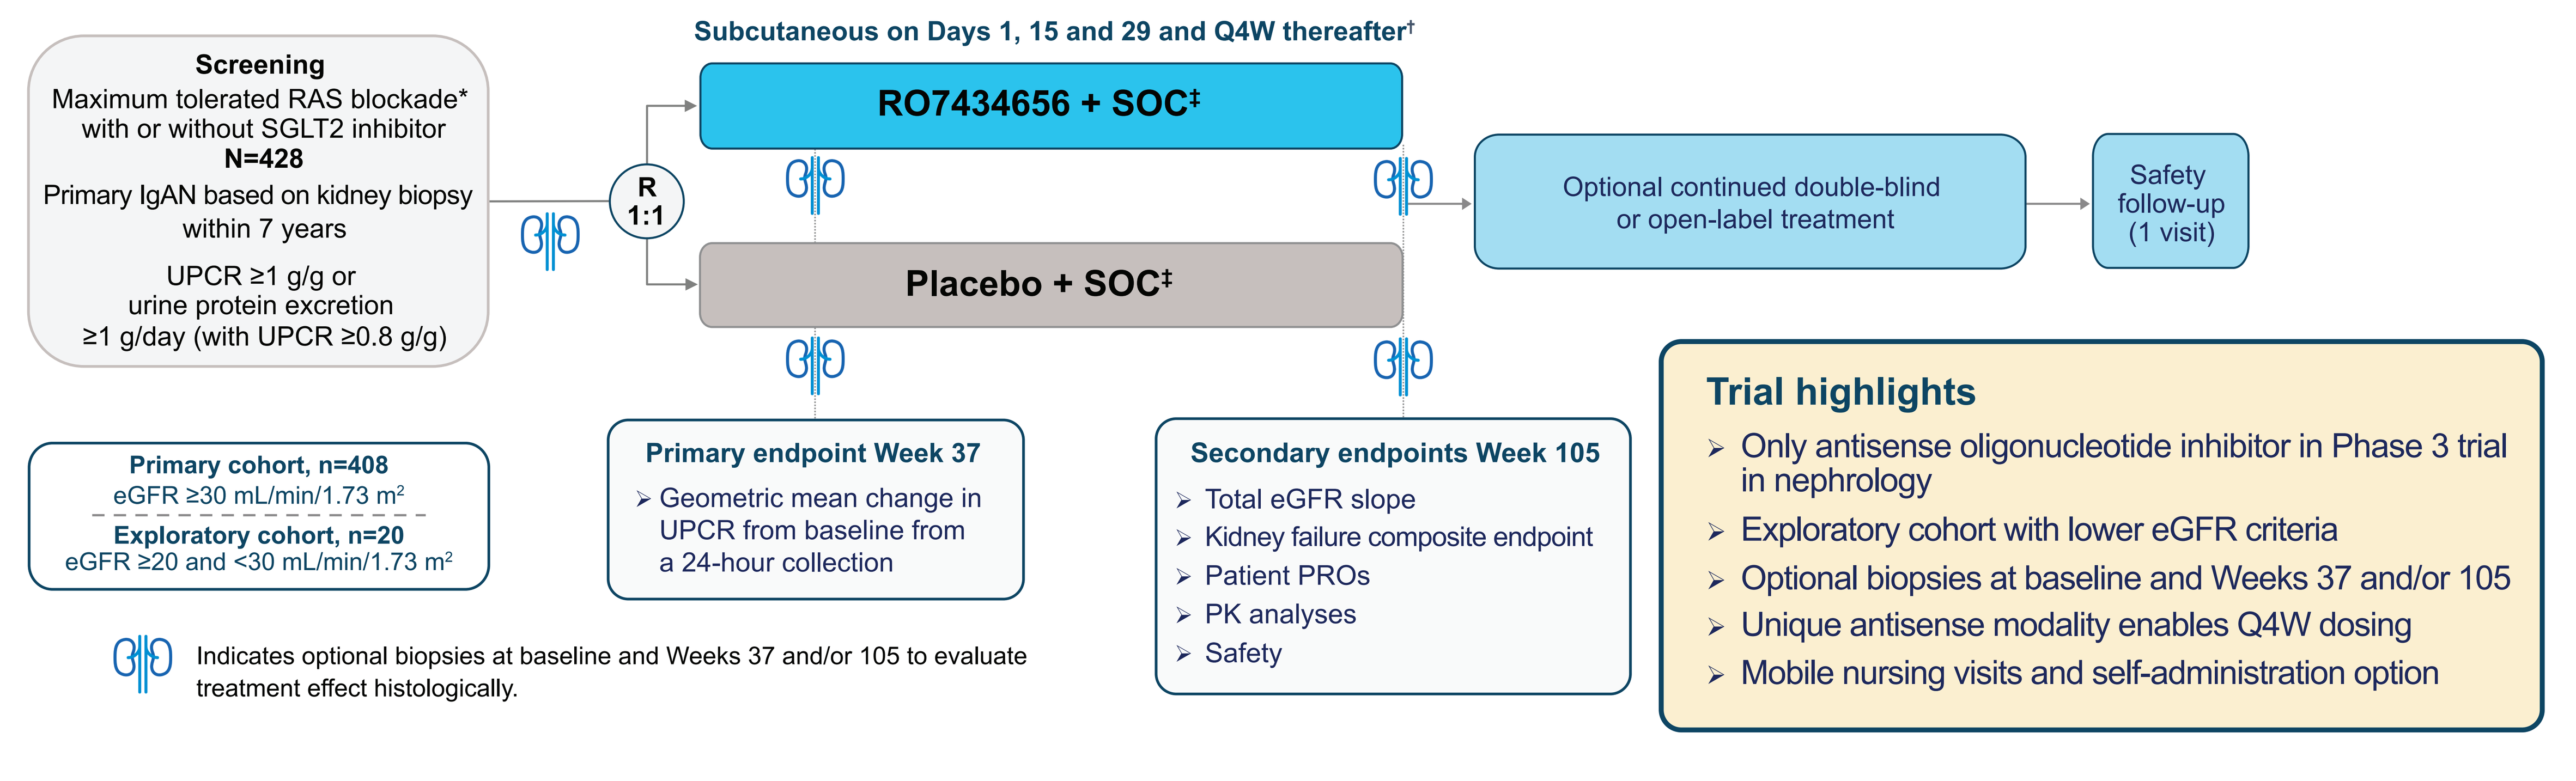

Screening
Maximum tolerated RAS blockade* with or without SGLT2 inhibitor
N=428
Primary IgAN based on kidney biopsy within 7 years
UPCR ≥1 g/g or urine protein excretion ≥1 g/day (with UPCR ≥0.8 g/g)
R 1:1
Subcutaneous on Days 1, 15 and 29 and Q4W thereafter†
RO7434656 + SOC‡
Placebo + SOC‡
Optional continued double-blind or open-label treatment
Safety follow-up (1 visit)
Primary cohort, n=408
eGFR ≥30 mL/min/1.73 m²
Exploratory cohort, n=20
eGFR ≥20 and <30 mL/min/1.73 m²
Primary endpoint Week 37
Geometric mean change in UPCR from baseline from a 24-hour collection
Secondary endpoints Week 105
Total eGFR slope
Kidney failure composite endpoint
Patient PROs
PK analyses
Safety
Indicates optional biopsies at baseline and Weeks 37 and/or 105 to evaluate treatment effect histologically.
Trial highlights
Only antisense oligonucleotide inhibitor in Phase 3 trial in nephrology
Exploratory cohort with lower eGFR criteria
Optional biopsies at baseline and Weeks 37 and/or 105
Unique antisense modality enables Q4W dosing
Mobile nursing visits and self-administration option

ACE, angiotensin-converting enzyme; ARB, angiotensin receptor blocker; BP, blood pressure; eGFR, estimated glomerular filtration rate; ERA, endothelin receptor antagonist; IgAN, IgA nephropathy; PK, pharmacokinetic; PRO, patient-reported outcome; Q4W, every 4 weeks; R, randomized; RAS, renin-angiotensin system; SGLT2, sodium glucose cotransporter 2; SOC, standard of care; UPCR, urine protein-to-creatinine ratio.
*Stable doses of SGLT2 inhibitors, ERAs, or other agent(s) for BP management permitted.
†Self-administration by participant or caregiver permitted after first 8 doses. Average on-site visits every 12 weeks, with the option of mobile nursing visits. Biomarker, blood and urine samples to be collected throughout the study.
‡SOC includes ACE inhibitor or ARB at maximum tolerated approved dose with or without SGLT2 inhibitors, ERAs or other agent(s) for BP management.

Figure 4. Enrollment Sites

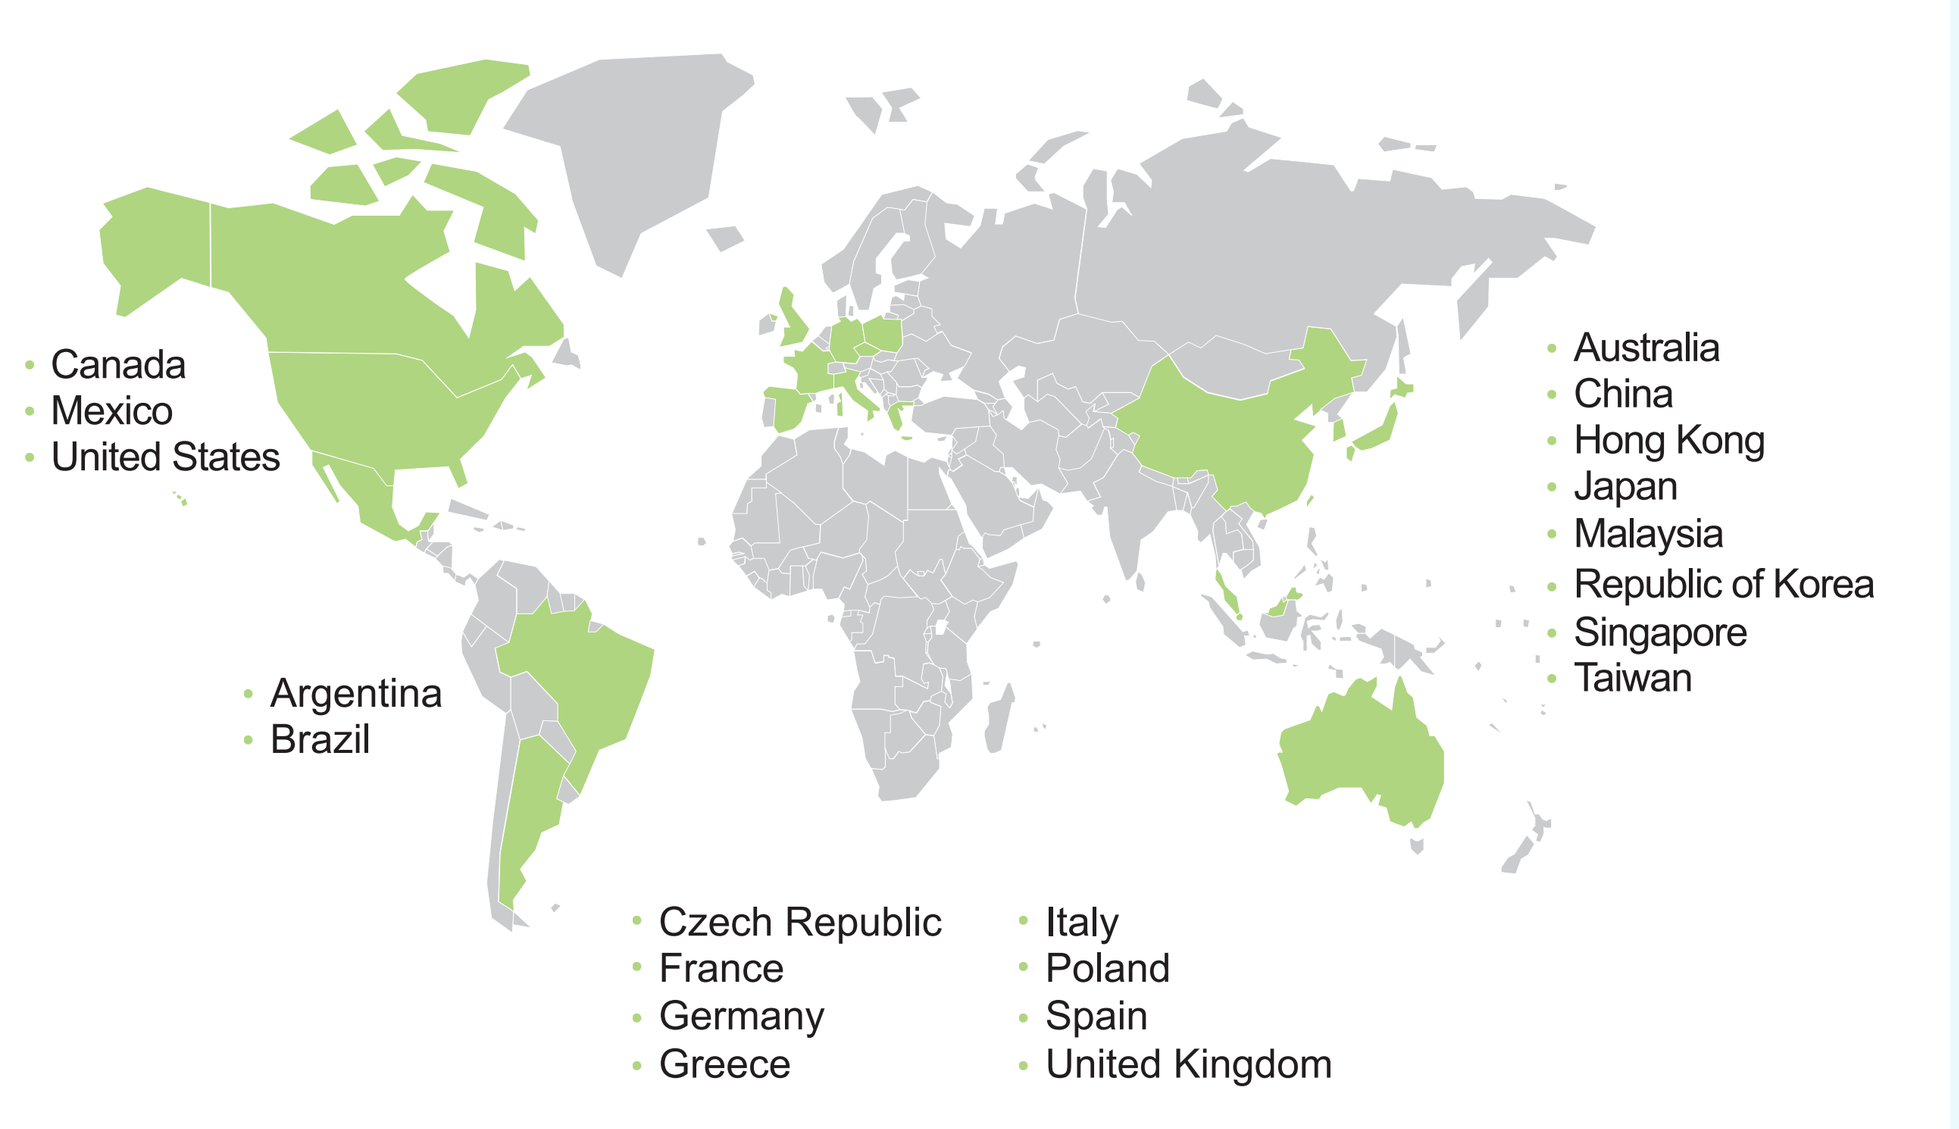

Canada
Mexico
United States
Argentina
Brazil
Czech Republic
France
Germany
Greece
Italy
Poland
Spain
United Kingdom
Australia
China
Hong Kong
Japan
Malaysia
Republic of Korea
Singapore
Taiwan

- Approximately 225 sites will participate in the study

Table 1. Key Inclusion and Exclusion Criteria

Key Inclusion Criteria	Key Exclusion Criteria
Diagnosed with primary IgAN as evidenced by a kidney biopsy performed within 7 years prior to or during screening Previous treatment with maximum tolerated doses of ACE inhibitors or ARBs for ≥90 days immediately prior to screening	Histopathologic or other evidence of another autoimmune glomerular disease Presence of ≥50% crescents in kidney biopsy, sustained doubling of serum creatinine level within 3 months prior to screening or rapidly progressive glomerulonephritis in the opinion of the investigator
UPCR ≥1 g/g or urine protein excretion ≥1 g/day (with UPCR ≥0.8 g/g) from a 24-hour collection eGFR ≥30 mL/min/1.73 m² (primary cohort)* or ≥20 and <30 mL/min/1.73 m² (exploratory cohort)*	Hemoglobin A1c ≥6.5% or a clinical diagnosis of diabetes mellitus of any type
Age ≥18 years at the time of signing of informed consent form Vaccination against *Neisseria meningitidis, Streptococcus pneumoniae and Haemophilus influenzae*†	Treatment with oral or IV corticosteroids with a dose equivalent to ≥7.5 mg/day of prednisone for 7 days or ≥5 mg/day of prednisone for 14 days within 90 days prior to screening Treatment with oral or IV corticosteroids during screening

ACE, angiotensin-converting enzyme; ARB, angiotensin receptor blocker; eGFR, estimated glomerular filtration rate; IgAN, IgA nephropathy; IV, intravenous; UPCR, urine protein-to-creatinine ratio.
*eGFR calculated by the 2021 Chronic Kidney Disease Epidemiology Collaboration creatinine equation.
†If vaccination must be administered during screening, it must occur ≥2 weeks prior to the first dose of RO7434656.

Table 2. Primary, Secondary and Exploratory Endpoints

Primary Endpoint
Change in proteinuria from baseline as measured by geometric mean change of **UPCR at Week 37** from a 24-hour collection
Secondary Endpoints
Total eGFR* slope at Week 105
Change in symptoms and health-related QOL at Week 105 compared with baseline as assessed by the Kidney Disease Quality of Life 36-Item Short Form survey
Time to first composite kidney failure endpoint†
Change in fatigue at Week 105 compared with baseline as assessed by the Functional Assessment of Chronic Illness Therapy–Fatigue Scale
Plasma concentrations of RO7434656
Incidence and severity of treatment-emergent adverse events
Exploratory Endpoints
Exploratory biomarker assessments

eGFR, estimated glomerular filtration rate; QOL, quality of life; UPCR, urine protein-to-creatinine ratio. *eGFR calculated using the 2021 Chronic Kidney Disease Epidemiology Collaboration creatinine equation.†Defined as receipt of kidney transplant, need for kidney replacement therapy or sustained decline in eGFR of ≥30%.

REFERENCES

1. Suzuki H, et al. *J Am Soc Nephrol.* 2011;22(10):1795-1803.
2. McGrogan A, et al. *Nephrol Dial Transplant.* 2011;26(2):414-430.
3. Kidney Disease: Improving Global Outcomes (KDIGO) Glomerular Diseases Work Group; Rovin BH, et al. *Kidney Int.* 2021;100(4 suppl):S1-S276.
4. van den Wall Bake AW, et al. *Am J Kidney Dis.* 1988;12(5):410-414.
5. Medjeral-Thomas NR, et al. *Semin Immunopathol.* 2021;43:679–690.
6. Barbour S, et al. Presented at: American Society of Nephrology (ASN) Kidney Week 2022; November 3-6, 2022; Orlando, FL. Abstract SA-PO714.

DISCLOSURES

J. Barratt has received consulting/speaker fees from Alnylam, Argenx, Astellas, BioCryst, Calliditas Therapeutics, Chinook, Dimerix, Galapagos, Novartis, Omeros, Travere Therapeutics, Vera Therapeutics and Visterra; and grant support from Argenx, Calliditas Therapeutics, Chinook, Galapagos, GSK, Novartis, Omeros, Travere Therapeutics and Visterra., **J. Flöge** has received consultancy and/or speaker honoraria from AstraZeneca, Boehringer, Calliditas, Chinook, Novartis, Omeros, Roche, Stadapharm, Travere and Vera Therapeutics. **V. Duggal** and **J. Lo** are employees of Genentech, Inc. **N. Schmit** and **J. Cheng** are employees of F. Hoffmann-La Roche Ltd. **B.H. Rovin** has received consulting fees from Roche/Genentech.

Ionis Pharmaceuticals discovered ASO Factor B and is partnered with F. Hoffmann-La Roche Ltd for its development.

Presented at: American Society of Nephrology (ASN) Kidney Week 2023; November 2-5, 2023; Philadelphia, PA.

This study is sponsored by F. Hoffmann-La Roche Ltd. Support for third-party writing assistance, provided by Samantha O'Dwyer, PhD, of Health Interactions, Inc., was funded by F. Hoffmann-La Roche Ltd.

An electronic version of this poster may be obtained by scanning this QR code with your smartphone app or go to the following URL: https://ter.li/7sqlea

More information on IMAGINATION may be obtained by scanning this QR code with your smartphone app or go to the following URL: https://ter.li/z7iyik

More information on the Barbour SJ, et al. ASN Kidney Week 2023 SA-PO926 poster (sponsored by Ionis) may be obtained by scanning this QR code with your smartphone app